Population pharmacokinetic analysis of avelumab in different cancer types

J. Wilkins[1], B. Brockhaus[2], S. Wang[2], H. Dai[3], B. Neuteboom[3], S. Brar[4], C. Bello[4], A. Khandelwal[2], P. Girard[5]

[1]Occams Coöperatie U.A., Amstelveen, The Netherlands; [2]Merck KGaA, Darmstadt, Germany; [3]EMD Serono Research & Development Institute, Inc, Billerica, MA, USA; [4]Pfizer, La Jolla, CA, USA; [5]Merck Institute for Pharmacometrics, Lausanne, Switzerland

BACKGROUND

- Avelumab is a human monoclonal antibody that selectively binds PD-L1, which is expressed on many different tumor cells
- Avelumab is approved in the US and EU for treatment of metastatic Merkel cell carcinoma (MCC), in Japan for curatively unresectable MCC, and platinum-treated advanced urothelial carcinoma (US),[1] and is in clinical development in other cancer types[2]
- Avelumab has shown promising clinical activity and manageable safety in multiple tumor types[2-6]
- Here, we present results from a population pharmacokinetic (PopPK) analysis accounting for covariates after multiple doses, considering models for time-varying clearance (CL) and covariate relationships

METHODS

Patients and treatment

- Pharmacokinetic and covariate data from three clinical trials were used for PopPK analysis:
 - JAVELIN Solid Tumor (NCT01772004): A Phase I, Open-label, Multiple-ascending Dose Trial to Investigate the Safety, Tolerability, Pharmacokinetics, Biological and Clinical Activity of Avelumab (MSB0010718C) in Subjects With Metastatic or Locally Advanced Solid Tumors and Expansion to Selected Indications (1,688 patients)
 - JAVELIN Solid Tumor JPN (NCT01943461): A Phase I Trial to Investigate the Tolerability, Safety, Pharmacokinetics, Biological and Clinical Activity of Avelumab (MSB0010718C) in Japanese Subjects With Metastatic or Locally Advanced Solid Tumors, With Expansion Part in Asian Subjects With Gastric Cancer (51 patients)
 - JAVELIN Merkel 200 (NCT02155647): A Phase II, Open-Label, Multicenter Trial to Investigate the Clinical Activity and Safety of Avelumab (MSB0010718C) in Subjects With Merkel Cell Carcinoma (88 patients)
- This analysis includes 10,637 avelumab serum concentration observations from 1,827 patients with 14 different tumor types, which were obtained according to rich and sparse sampling schemes
- Patients received avelumab at 1 mg/kg (n=4), 3 mg/kg (n=18), 10 mg/kg (n=1,778) or 20 mg/kg (n=27) every 2 weeks (Q2W) by intravenous (IV) infusion administered over 1 hour

Modeling

- The PopPK model was built using nonlinear mixed effect modeling software package (NONMEM; v7.3.0)
- Two-compartment models with covariates including time-constant and time-varying clearance (CL) were tested
- Changes in CL over time were considered using the model according to Liu et al.[7]:

$$CL = TVCL \bullet \exp\left(\frac{I_{max} \cdot Time^{\gamma}}{T50^{\gamma} + Time^{\gamma}}\right)$$

where *CL* is clearance (L/hour); *TVCL* is CL at baseline (L/hour); I_{max} is the logarithm of maximal change in CL relative to baseline; *T50* is time required to reach 50% of I_{max} (days); ɣ is shape parameter; and *Time* (days)

- A full-model approach was used for inclusion of covariates in the model[8]
- Covariates were tested on CL and central volume of distribution (V_1)
- Parameters controlling the time variability of CL were assessed individually by tumor type
- Categorical variables were implemented using a linear function, as follows:

$$PARCOV_i = PAR \bullet (1+\theta_{COV})$$

where *PARCOVi* is the parameter value for individual *i*; *PAR* is the typical value of the parameter in the population; and θ_{COV} is an estimated parameter corresponding to the unique value of the categorical covariate in individual *i*; for the largest category, θ_{COV} is defined as 0

- Continuous variables were defined by a power function, as follows:

$$PARCOV_i = PAR \bullet \left(\frac{COV_i}{\overline{COV}}\right)^{\theta COV}$$

where *PARCOVi* and *PAR* are as previously defined; COV_i is the value of the covariate in individual *i*; ($\overline{COV}$) is the median value of the covariate in the population; and θ_{COV} is a parameter describing the shape of the relationship of the covariate to the parameter

- Alternatively, time-varying covariates were included in the two-compartment model for CL according to Wählby et al.[9]:

$$CLCOV_{i,t} = TVCL \bullet [1+\theta_{BCOV} \bullet (COV_{base,i} - \overline{COV_{base}}) + \theta_{DCOV} \bullet (COV_{i,t} - COV_{base,i})]$$

where $CLCOV_{i,t}$ is the value of the CL in individual *i* at time *t*; *TVCL* is the typical value of CL in the population; θ_{BCOV} is a parameter describing the proportional relationship of the normalized covariate at baseline to the parameter; $COV_{base,i}$ is the value of the covariate at baseline in individual *i*; $\overline{COV_{base}}$ is the median value of the covariate in the population at baseline; θ_{DCOV} is a parameter describing the relationship between the parameter and change from baseline in the covariate; and $COV_{i,t}$ is the value of the covariate in individual *i* at time *t*

- The following time-varying covariates were included in the model, each separately:
 - Body weight, albumin, platelet count, C-reactive protein, and tumor size

RESULTS

- Avelumab concentrations increased over time
- Incorporation of time-varying covariates did not result in model improvement, as judged by plots of conditional-weighted residuals against time (**Figure 1**) as well as by decrease in Objective Function Value
- A two-compartment model incorporating time-varying CL provided the best description of avelumab concentration (**Table 1**, **Figure 2**, **Figure 3**)
- Significant decreases in CL over time were seen in patients with MCC or head & neck cancer, reaching a maximal decrease of 32.1% and 24.7%, respectively (**Table 1**, **Figure 4**)
- No significant decreases in CL over time were seen in any of the other tumor types represented in the dataset
- In patients with MCC, this may reflect that they were typically followed longer than patients in other indications
- It could also be shown that time-varying CL is more prevalent in responders than non-responders
- Other significant covariates on CL included body weight (estimated exponent=0.324), albumin, tumor burden, age, sex, race, estimated glomerular filtration rate (eGFR), immunogenicity, platelet count, aspartate aminotransferase (AST) levels, C-reactive protein (CRP), concomitant opioid use, and previous use of biologics (**Figure 4**)
- For V_1, baseline body weight was of potential clinical importance (estimated exponent=0.362), with sex, albumin, and previous use of biologics producing small but statistically significant effects (**Figure 4**)

Figure 1. Conditional weighted residuals vs time after inclusion of the effect of time-varying tumor size on CL

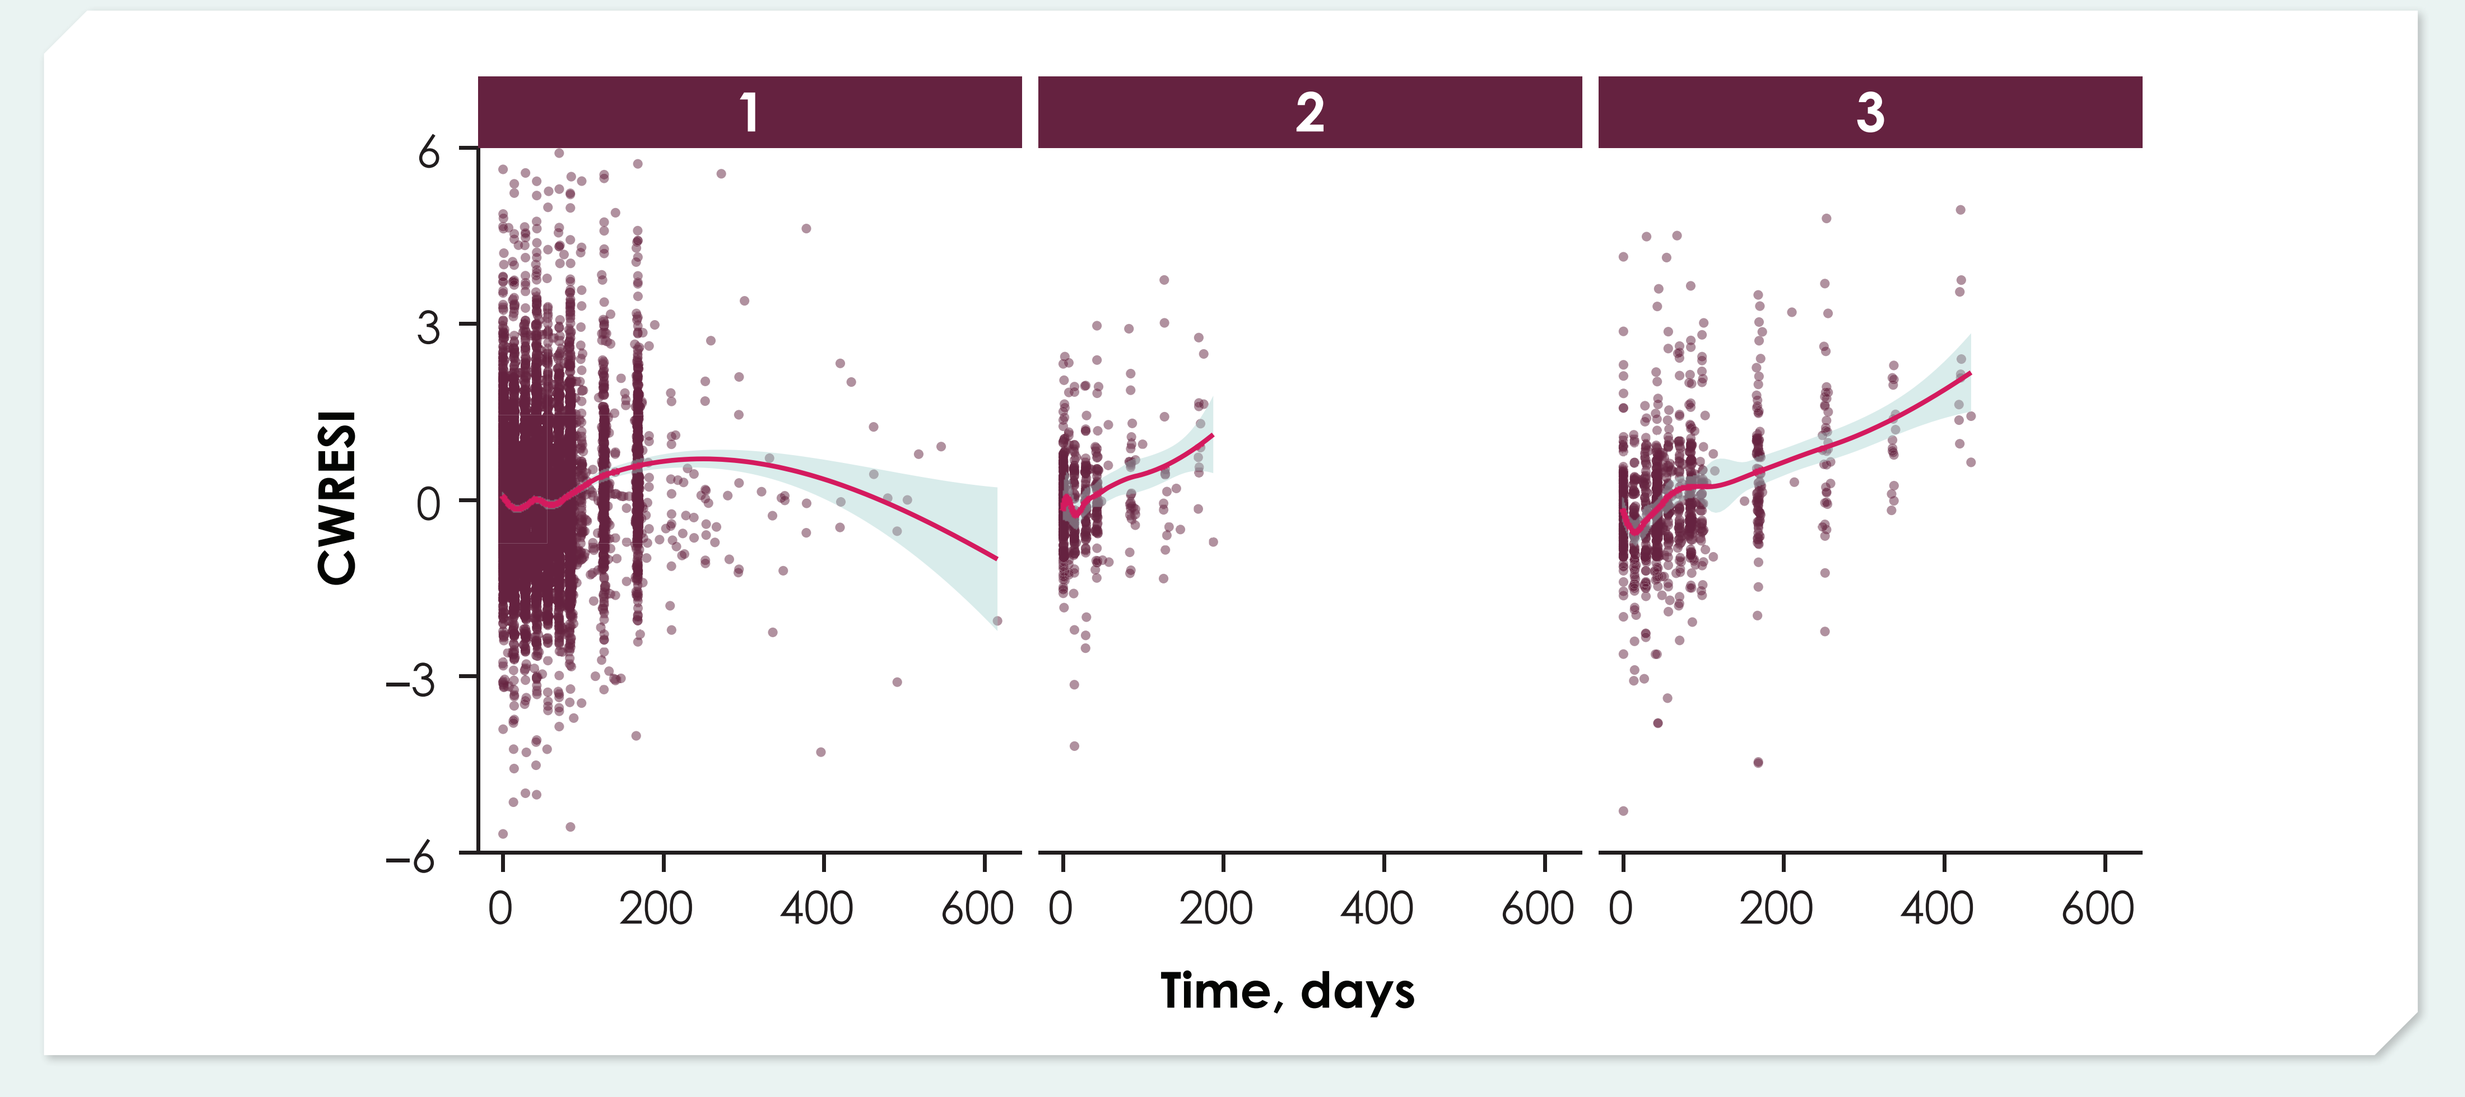
1
2
3
CWRESI
6
3
0
−3
−6
0
200
400
600
Time, days

CWRESI, conditional weighted residuals with full interaction
Conditioned by study: 1=JAVELIN Solid Tumor, 2=JAVELIN Solid Tumor JPN, 3=JAVELIN Merkel 200. Points are observations. Orange lines are loess smooths.

Table 1. Final reduced model (including time-varying CL with IIV on I_{max})

Parameter	Estimate	%RSE*	95% CI†	Shrinkage
Clearance (TVCL), L/hour	0.0308	1.36	0.0300, 0.0316	
Central volume (TVV_1), L	3.42	1.20	3.34, 3.50	
Peripheral volume (TVV_2), L	0.918	6.75	0.796, 1.04	
Intercompartmental clearance (TVQ), L/hour	0.0313	5.92	0.0277, 0.0349	
Max change in CL relative to baseline (θI_{max},ref, reference)	0‡	–	–	
Max change in CL relative to baseline ($\theta_{Imax,HN}$, TT: head & neck)	-0.284	18.8	-0.389, -0.18	
Max change in CL relative to baseline (θ_{Imax},MCC, TT: MCC)	-0.387	15.9	-0.507, -0.266	
Time required to reach 50% of Imax ($\theta_{T50,ref}$,reference), days	68.4	2.54	65, 71.8	
IIV				
IIV on CL (ω^2_{CL}, variance)	0.0535	6.71	0.0464; 0.0605	19.7
cov(CL,V_1) ($\omega_{CL,V2}$, covariance)	0.0171	13.8	0.0125; 0.0218	
IIV on V_1 (, variance)	0.0332	5.31	0.0297; 0.0366	36.3
cov(CL,V_2) (, covariance)	0.0633	25.2	0.0321; 0.0946	
cov(V_1,V_2) ($\omega_{CL,V2}$, covariance)	-0.014	-105	-0.0429; 0.0148	
IIV on V_2 (ω^2_{V2}, variance)	0.858	11.8	0.659; 1.06	54.7
IIV on I_{max} (ω^2_{Imax}, variance)	0.0596	9.51	0.0485; 0.0707	38.1
Residual variability				
Proportional residual error ($\sigma prop$)	0.162	0.521	0.161; 0.164	13.5
Additive residual error (σadd), (μg/mL)	2.43	0.904	2.39; 2.48	

IIV, interindividual variability; **TT**, tumor type
Covariate-PK parameter relationship not included
* %RSE = SE / estimate * 100
† Calculated as estimate ± 1.96 · SE
‡ Fixed

Figure 2. Conditional weighted residuals for the final reduced model

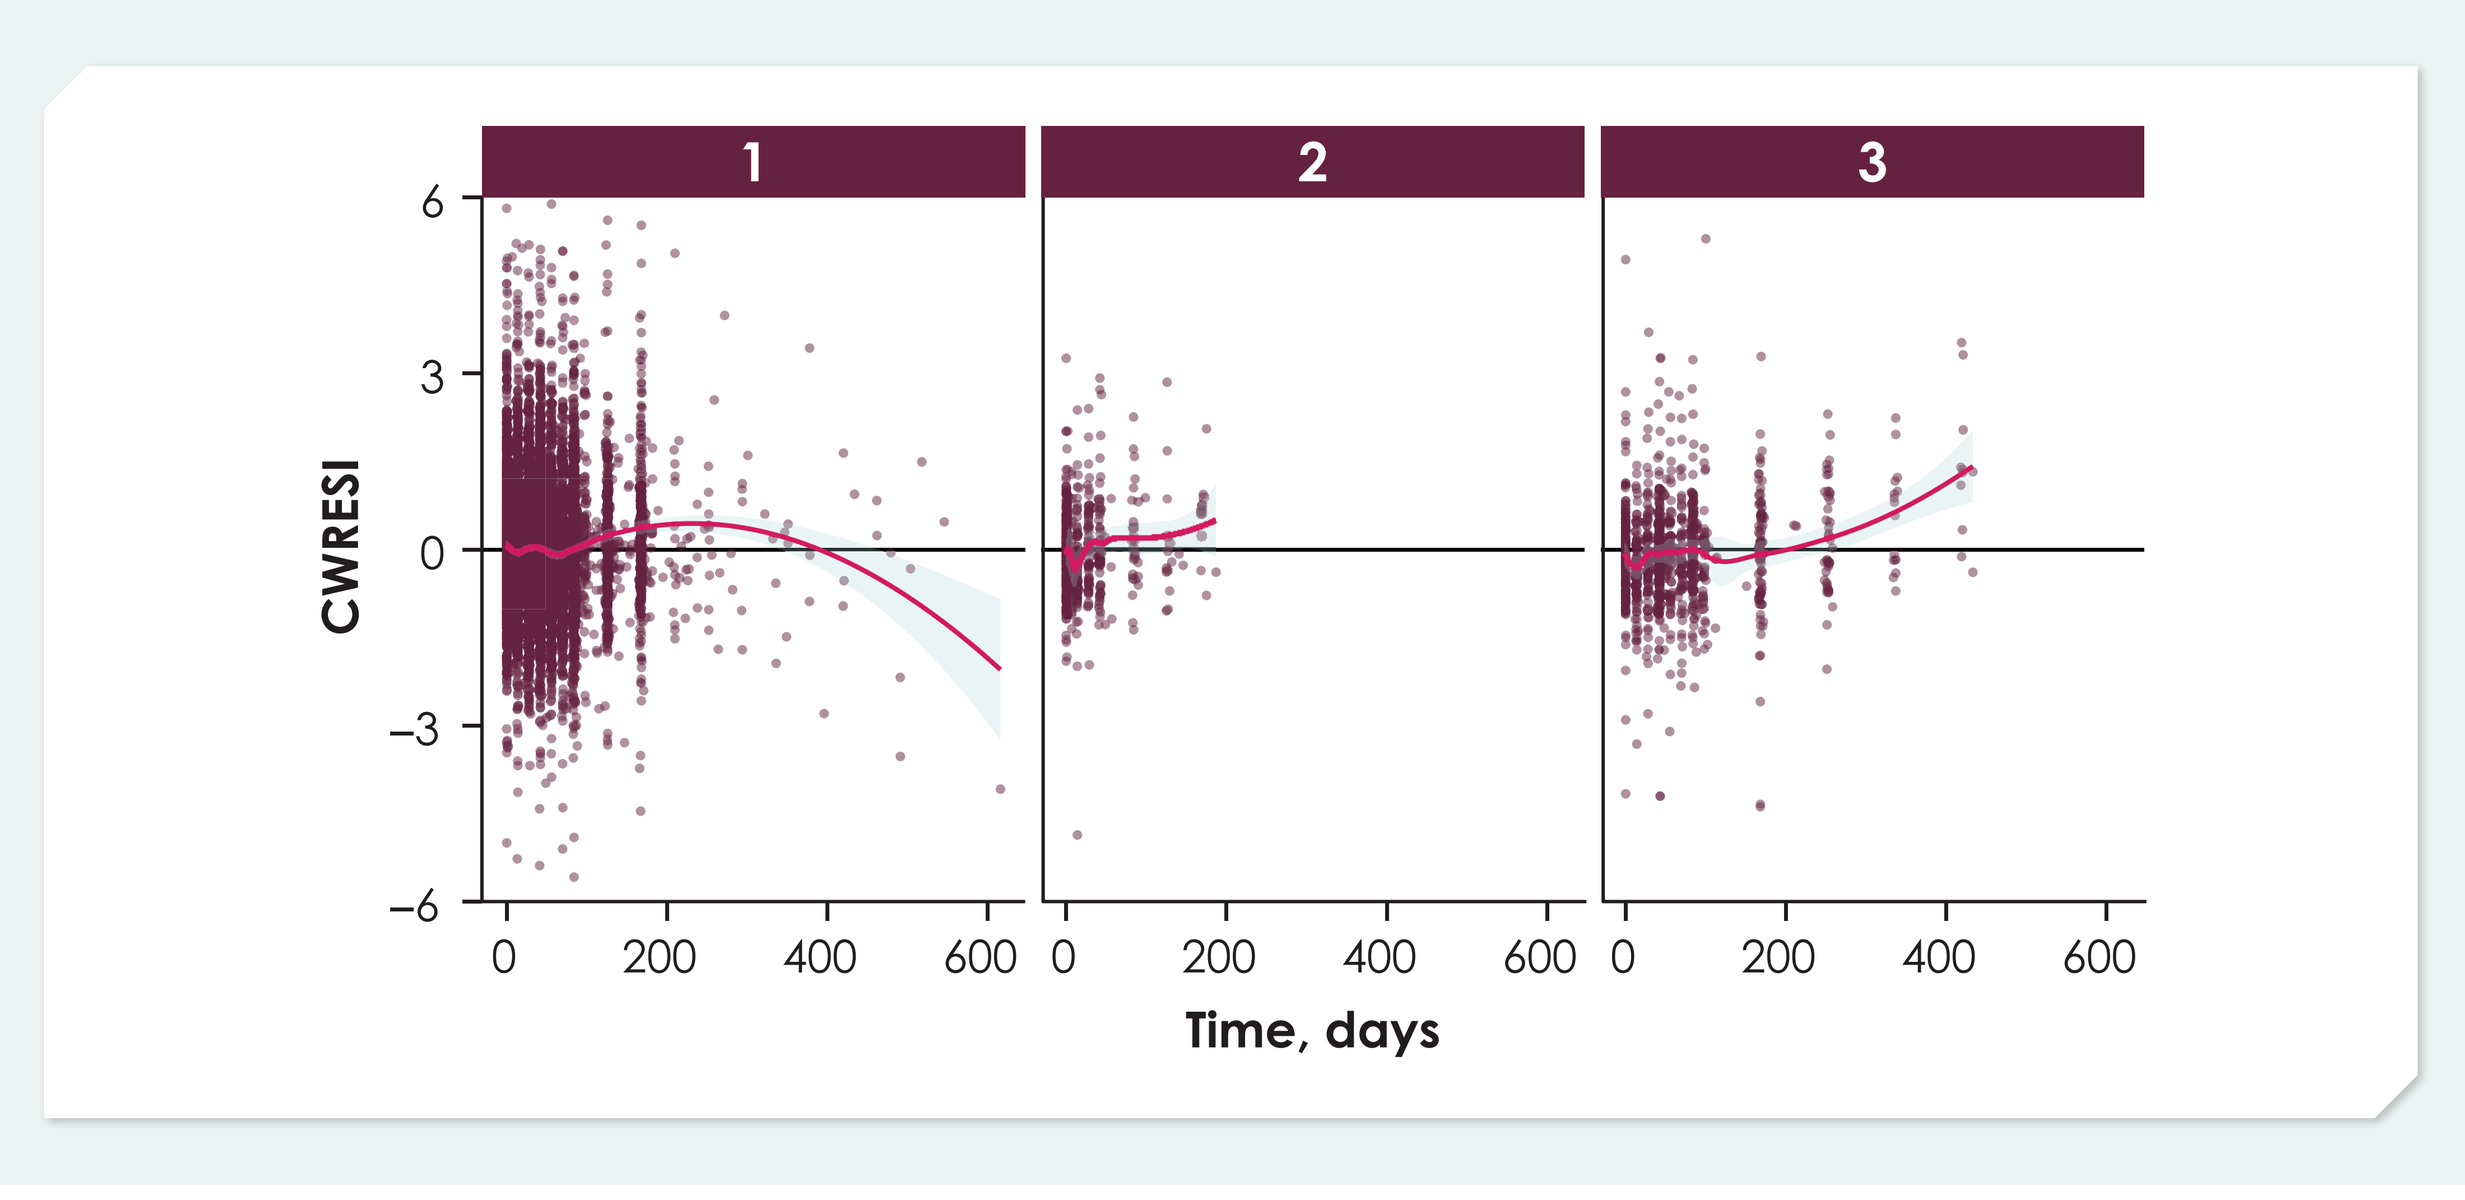
1
2
3
CWRESI
6
3
0
−3
−6
0
200
400
600
Time, days

Conditioned by study: 1=JAVELIN Solid Tumor, 2=JAVELIN Solid Tumor JPN, 3=JAVELIN Merkel 200. Points are observations. Orange lines are loess smooths through the data. Black lines are lines of identity or zero lines as appropriate.

Figure 3. Visual predicted check for the final reduced model

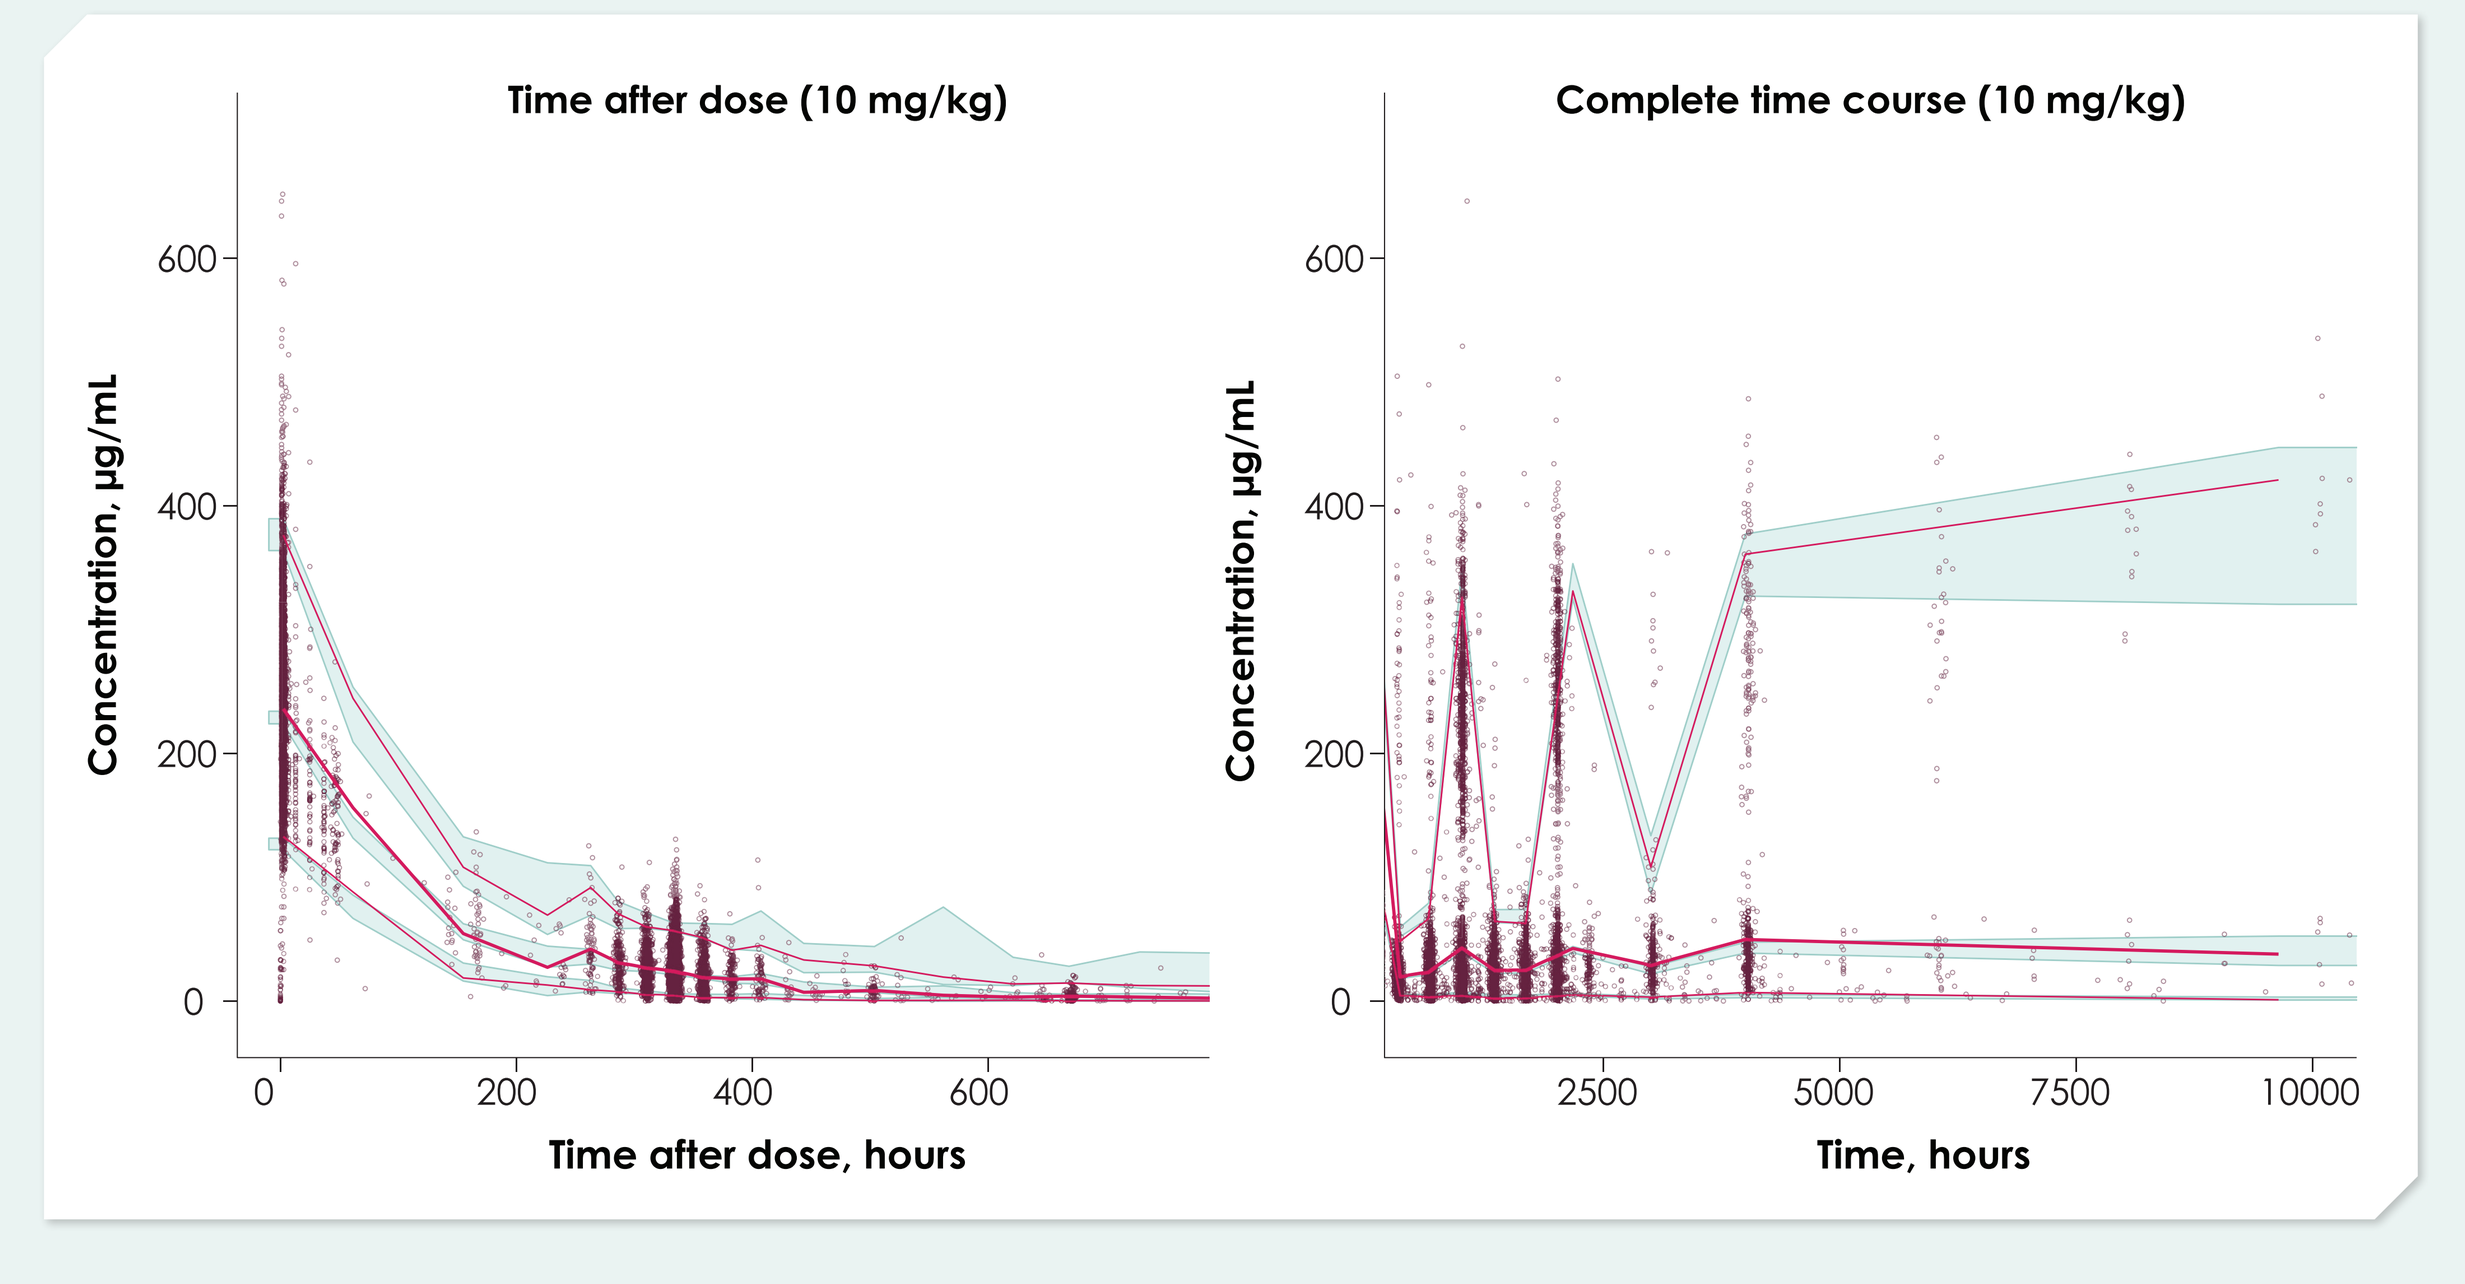
Time after dose (10 mg/kg)
Concentration, μg/mL
600
400
200
0
200
400
600
Time after dose, hours
Complete time course (10 mg/kg)
Concentration, μg/mL
2500
5000
7500
10000
Time, hours

Red points are observations. Blue lines are observed median, 5th and 95th percentiles. Orange shaded areas are 95% confidence intervals for simulated median, 5th and 95th percentiles.

Figure 4. Relationships between covariates and PK parameters (CL, V_1 and I_{max})

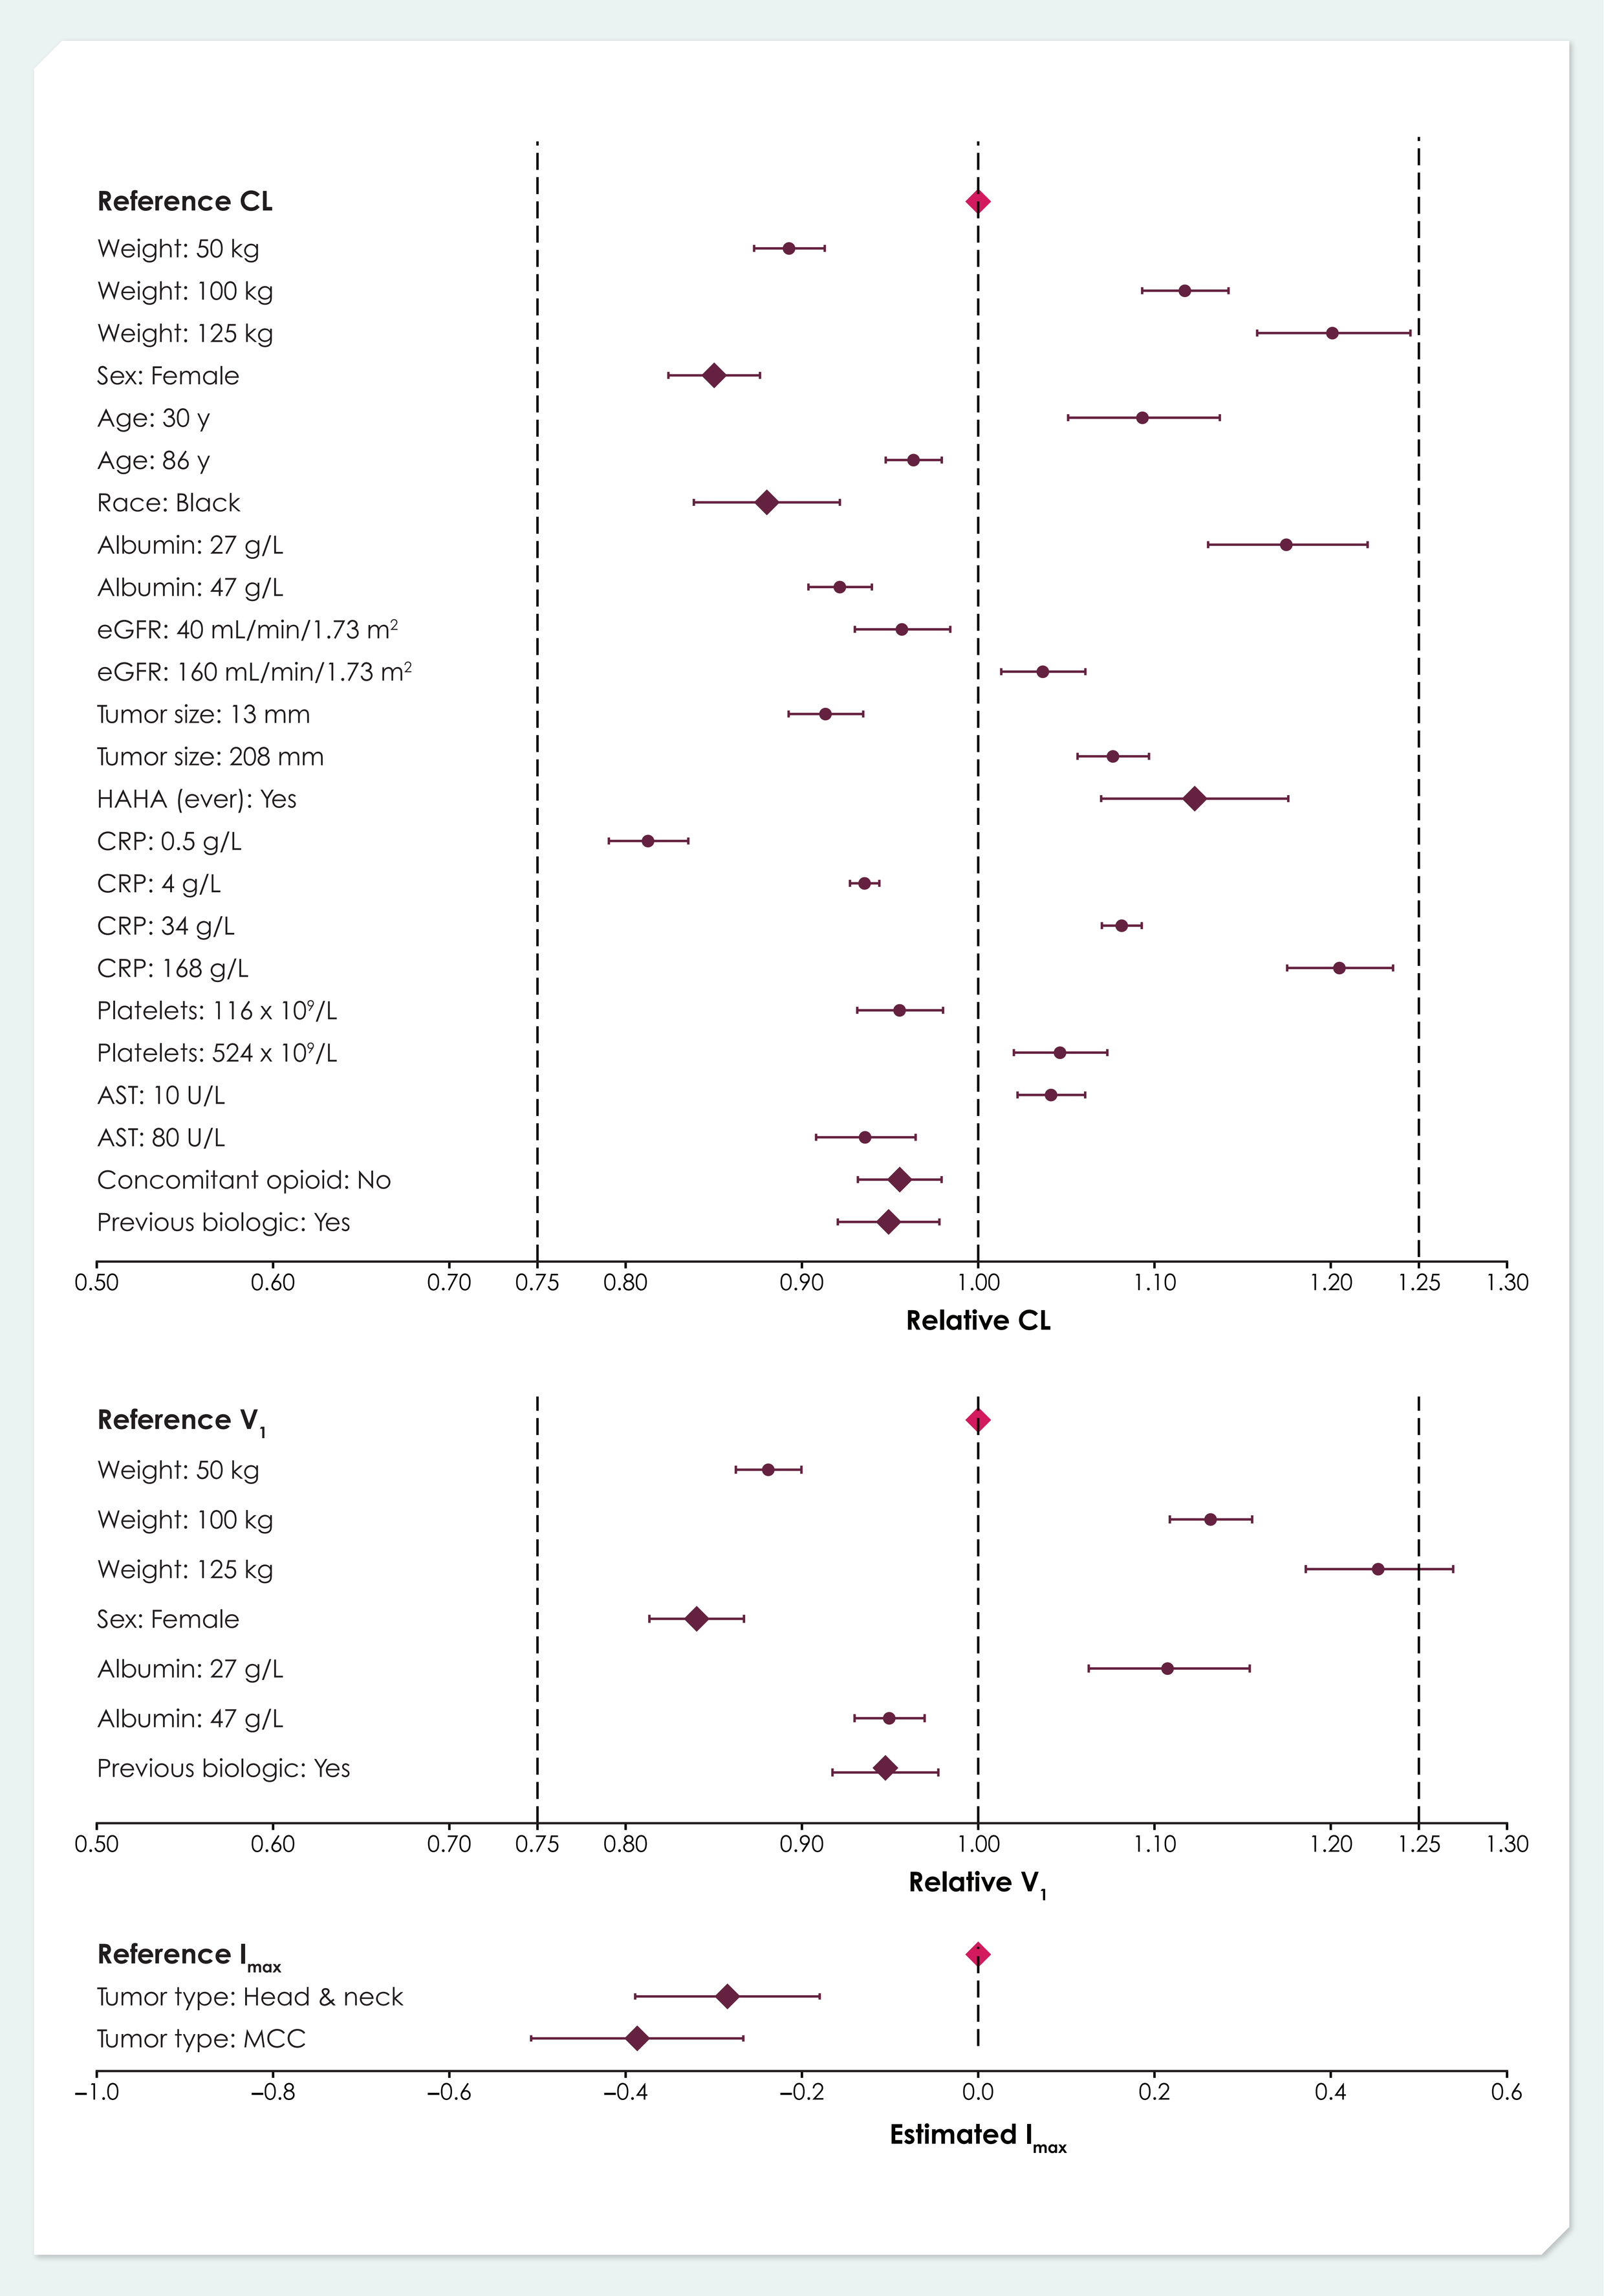
Reference CL
Weight: 50 kg
Weight: 100 kg
Weight: 125 kg
Sex: Female
Age: 30 y
Age: 86 y
Race: Black
Albumin: 27 g/L
Albumin: 47 g/L
eGFR: 40 mL/min/1.73 m²
eGFR: 160 mL/min/1.73 m²
Tumor size: 13 mm
Tumor size: 208 mm
HAHA (ever): Yes
CRP: 0.5 g/L
CRP: 4 g/L
CRP: 34 g/L
CRP: 168 g/L
Platelets: 116 x 10⁹/L
Platelets: 524 x 10⁹/L
AST: 10 U/L
AST: 80 U/L
Concomitant opioid: No
Previous biologic: Yes
0.50
0.60
0.70
0.75
0.80
0.90
1.00
1.10
1.20
1.25
1.30
Relative CL
Reference V1
Weight: 50 kg
Weight: 100 kg
Weight: 125 kg
Sex: Female
Albumin: 27 g/L
Albumin: 47 g/L
Previous biologic: Yes
Relative V1
Reference Imax
Tumor type: Head & neck
Tumor type: MCC
−1.0
−0.8
−0.6
−0.4
−0.2
0.0
0.2
0.4
0.6
Estimated Imax

HAHA, human anti-human antibody
Small diamonds with error bars represent effects of continuous covariates at specific covariate values of clinical interest. Large diamonds with error bars represent effects of categorical covariates with 95% confidence intervals. Vertical dashed lines represent relative CL values of 0.75, 1, and 1.25.

CONCLUSIONS

- **The PopPK model incorporating time-varying CL described the observed data well**
- **Time-varying CL has also been observed in other anti–PD-1/PD-L1 antibodies such as nivolumab, pembrolizumab and atezolizumab[7,10,11]**
- **It may be related to post-treatment effects on disease impacting exposure, such as inflammation, cachexia, tumor dynamics, or treatment efficacy[7]**
- **The identified covariates did not warrant dose adjustment**

REFERENCES

1. Bavencio (avelumab) [package insert]. Darmstadt, Germany: Merck KGaA; 2017.
2. Heery CR, et al. Lancet Oncol. 2017;18(5):587-97.
3. Kaufman HL, et al. Cancer Res. 2017;77(13 suppl):Abstract CT079.
4. Patel M, et al. J Clin Oncol. 2017;35(suppl 6S):Abstract 330.
5. Gulley JL, et al. Lancet Oncol. 2017;18(5):599-610.
6. Jerusalem G, et al. J Thoracic Oncol. 2017;12(S1):Abstract OA03.03.
7. Liu C, et al. Clin Pharmacol Ther. 2017;101(5):657-66.
8. Gastonguay M. PAGE 20, Athens, Greece, 2011:Abstract 2229.
9. Wählby U, et al. Br J Clin Pharmacol. 2004;58(4):367-77.
10. Li H, et al. J Pharmacokinet Pharmacodyn. 2017 Jun 1. [Epub ahead of print].
11. Atezolizumab. https://www.accessdata.fda.gov/drugsatfda_docs/nda/2016/761041Orig1s000ClinPharmR.pdf.

ACKNOWLEDGMENTS

The authors thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers and at Merck KGaA, Darmstadt, Germany, and EMD Serono, Billerica, MA, USA (a business of Merck KGaA, Darmstadt, Germany). The trials reported in this analysis were sponsored by Merck KGaA, Darmstadt, Germany, and are part of an alliance between Merck KGaA, Darmstadt, Germany and Pfizer Inc, New York, NY, USA. Medical writing support was provided by ClinicalThinking Inc, Hamilton, NJ, USA, and funded by Merck KGaA, Darmstadt, Germany, and Pfizer Inc, New York, NY, USA.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

Correspondence: Justin Wilkins, justin.wilkins@occams.com

GET POSTER PDF

Poster presentation at the 8th ACoP Annual Meeting, October 15-18, 2017; Marriott Resort: Harbor Beach Hotel, Fort Lauderdale, FL, USA