Adverse event management during treatment with bintrafusp alfa, a bifunctional fusion protein targeting TGF-β and PD-L1: treatment guidance based on experience in clinical trials

Copies of this e-poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

GET POSTER PDF

J. L. Gulley[1], M. Lacouture[2], A. Spira[3,4], H. Verdaguer Mata[5], C. Yoo[6], B. C. Cho[7], C. Helwig[8], T. Halady[8], C. Valencia[9], M. Bajars[8], J. Strauss[10], I. Brownell[11]

[1]Genitourinary Malignancies Branch, Center for Cancer Research, National Cancer Institute, National Institutes of Health, Bethesda, MD, USA; [2]Dermatology Service, Memorial Sloan Kettering Cancer Center, New York, NY, USA; [3]Virginia Cancer Specialists, Fairfax, VA, USA; [4]US Oncology Research, The Woodlands, TX, USA; [5]Hospital Universitari Vall d'Hebron, Barcelona, Spain; [6]Asan Medical Center, Seoul, Republic of Korea; [7]Yonsei Cancer Center, Yonsei University College of Medicine, Seoul, Republic of Korea; [8]Merck Healthcare KGaA, Darmstadt, Germany; [9]EMD Serono Research & Development Institute, Inc., Billerica, MA, USA, an affiliate of Merck KGaA; [10]Laboratory of Tumor Immunology and Biology, Center for Cancer Research, National Cancer Institute, National Institutes of Health, Bethesda, MD, USA; [11]Cutaneous Development and Carcinogenesis Section, National Institute of Arthritis and Musculoskeletal and Skin Diseases, National Institutes of Health, Bethesda, MD, USA

INTR@PID CLINICAL TRIALS™

BACKGROUND

- Bintrafusp alfa demonstrated clinical activity and manageable safety in patients with heavily pretreated solid tumors from the two phase 1 studies, INTR@PID 001 (NCT02517398) and INTR@PID 008 (NCT02699515)
- Bintrafusp alfa AEs of special interest included TGF-β inhibition–mediated skin AEs, irAEs, anemia, bleeding events, and IRRs
- Bintrafusp alfa-associated AEs observed to date were mostly mild to moderate and effective management will be essential to minimize treatment interruptions and to maintain patients' QOL

Proposed mechanism of action of bintrafusp alfa

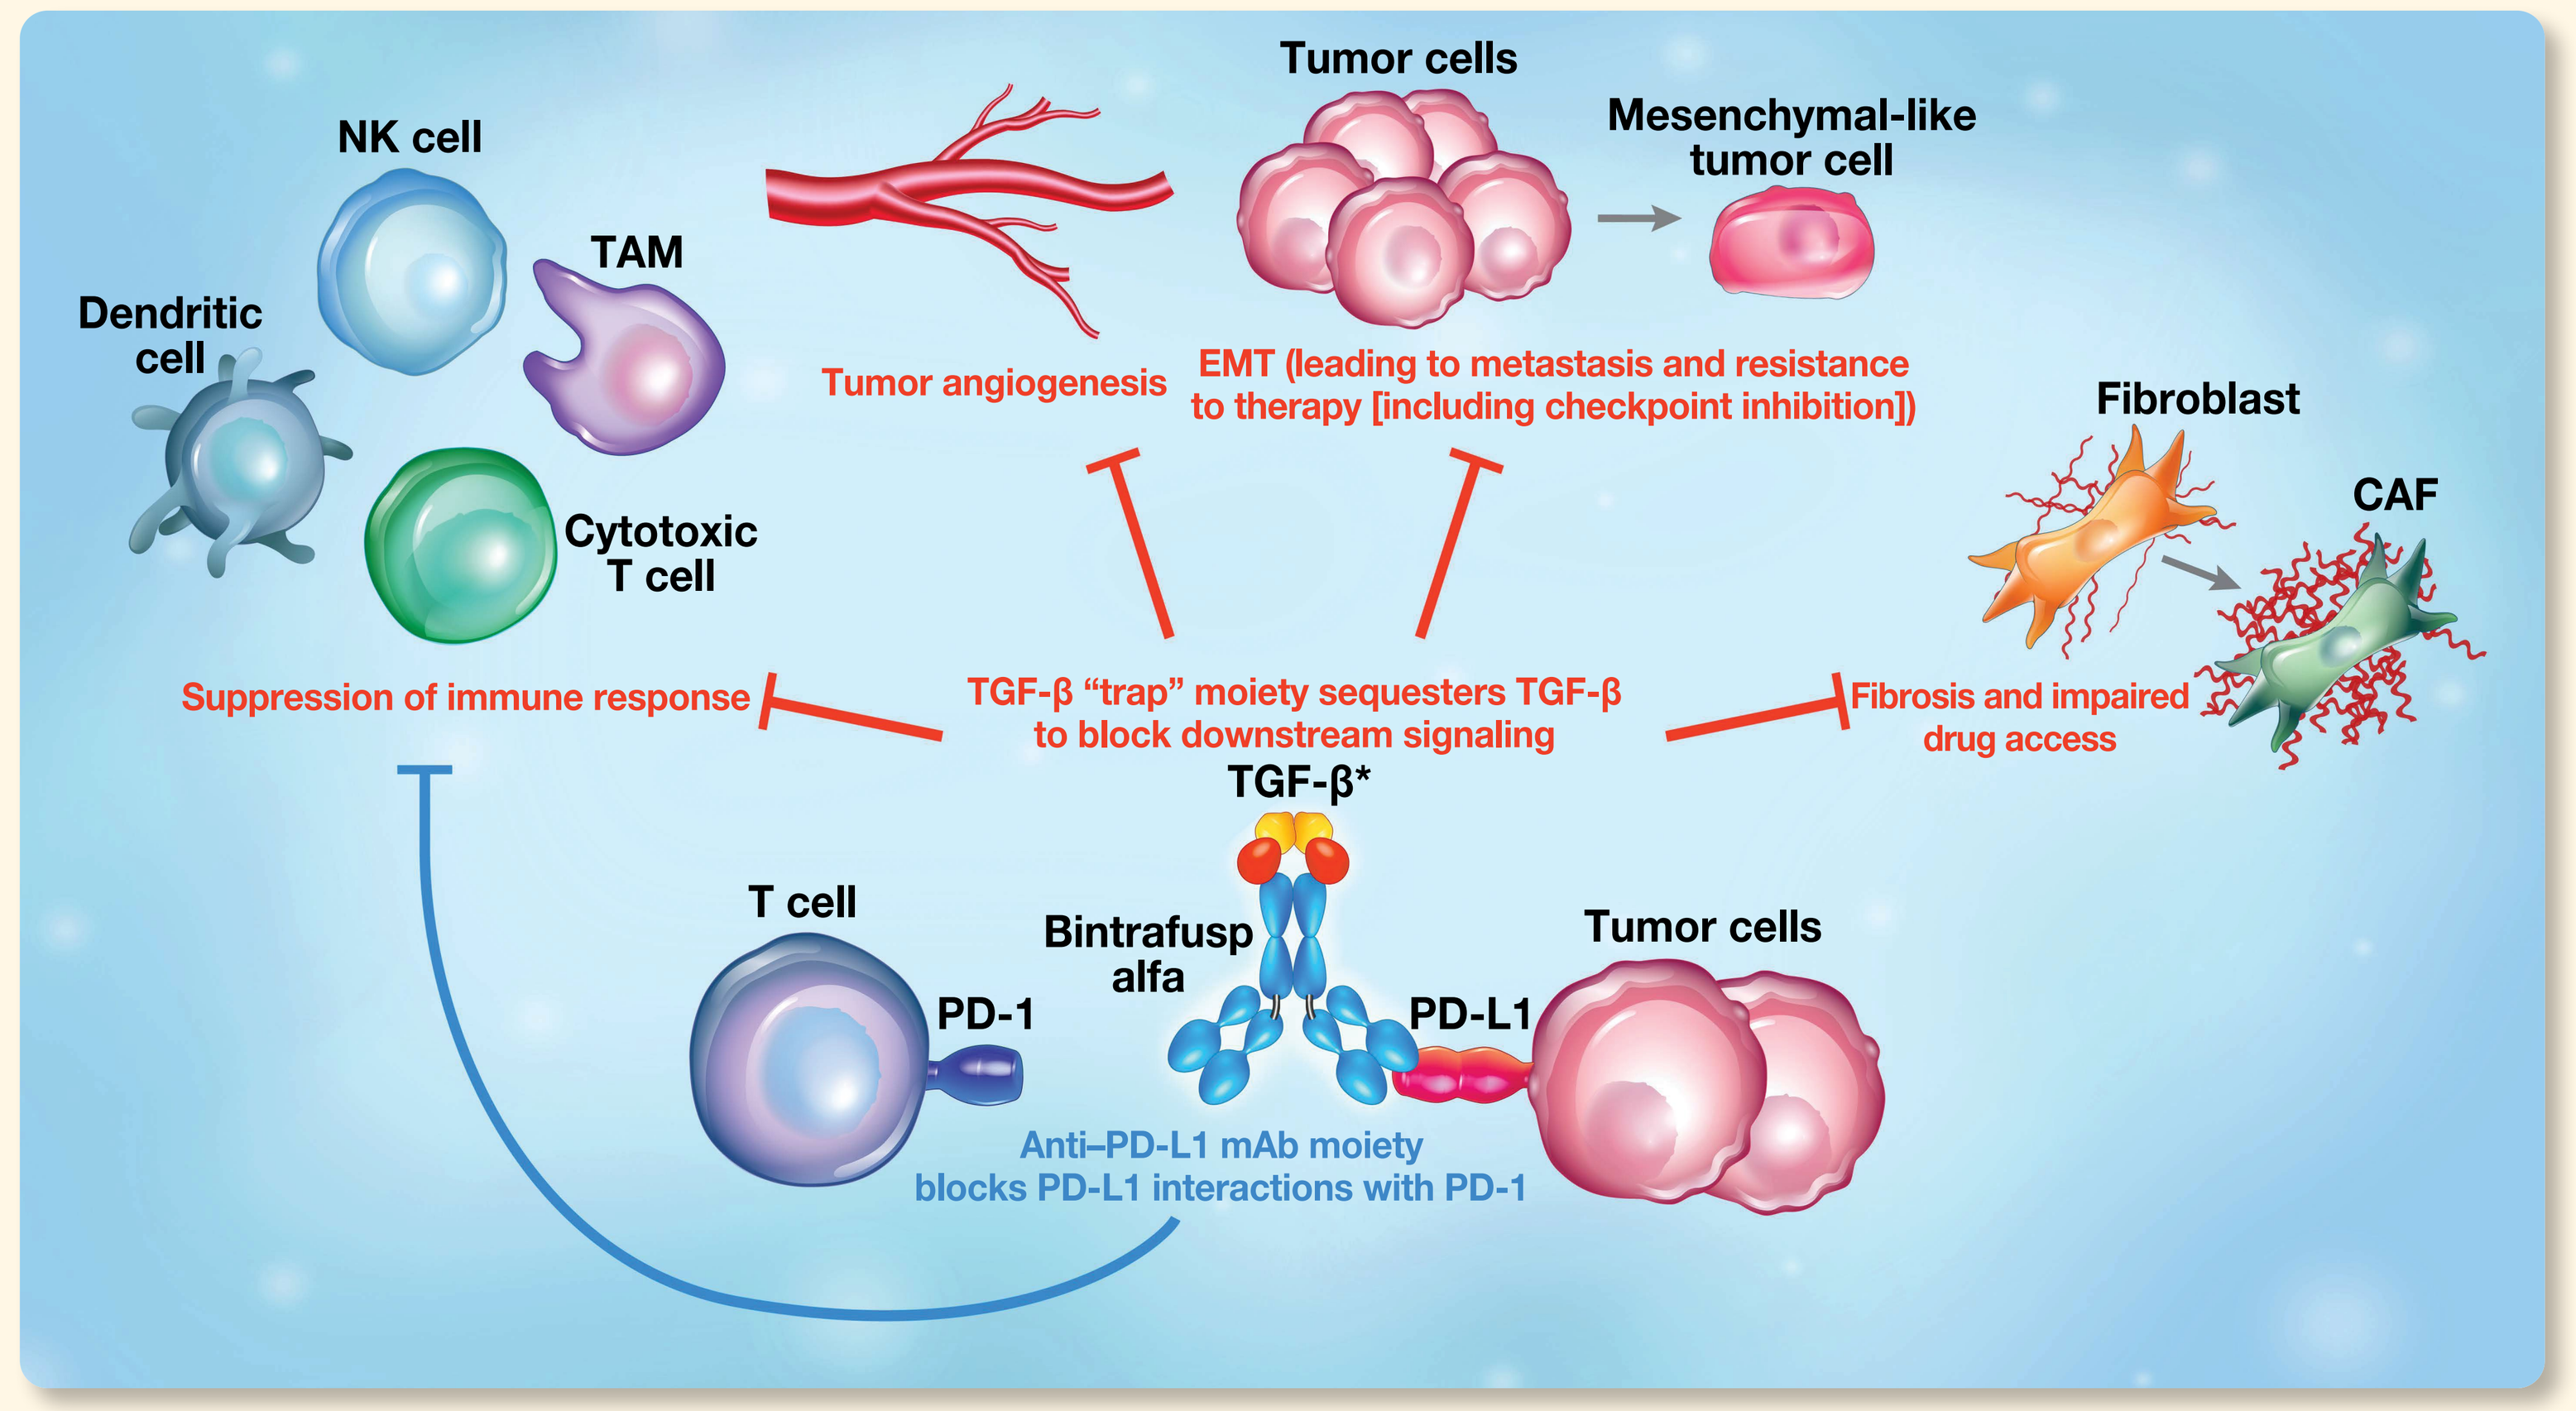
Tumor cells
Mesenchymal-like tumor cell
NK cell
TAM
Dendritic cell
Cytotoxic T cell
Tumor angiogenesis
EMT (leading to metastasis and resistance to therapy [including checkpoint inhibition])
Fibroblast
CAF
Suppression of immune response
TGF-β "trap" moiety sequesters TGF-β to block downstream signaling
Fibrosis and impaired drug access
TGF-β*
T cell
Bintrafusp alfa
Tumor cells
PD-1
PD-L1
Anti–PD-L1 mAb moiety blocks PD-L1 interactions with PD-1

*Tumor cells are also a major source of TGF-β in the microenvironment.

- Bintrafusp alfa is a first-in-class bifunctional fusion protein composed of the extracellular domain of the TGF-βRII receptor to function as a TGF-β "trap" fused to a human IgG1 antibody blocking PD-L1[1,2]
- The bifunctional nature of bintrafusp alfa might allow for simultaneous, colocalized inhibition of two nonredundant immunosuppressive pathways, TGF-β and PD-L1, within the TME

METHODS

- We analyzed AEs observed in 606 patients receiving a 1200 mg dose of bintrafusp alfa in the phase 1 INTR@PID 001 and 008 studies
- With the aim of optimizing patients' treatment experience and outcomes, we provide guidance for the early identification and management of these AEs which may help to mitigate the impact of bintrafusp alfa–associated AEs on patients and potentially increase treatment duration

RESULTS

- Most patients were male (67.3%), were aged <65 years (59.6%), and had received ≥2 prior anticancer regimens (58.3%)
- The most common primary tumor type was NSCLC (27.1%); other primary tumor types included HCC (17.5%), BTC (5.0%), and cervical (2.5%)
- TRAEs of any grade occurred in 68.3% of patients (n=414), and TRAEs grade ≥3 occurred in 22.3% of patients (n=135); 8.7% of patients (n=53) permanently discontinued treatment due to TRAEs

View baseline characteristics

View safety overview

TGF-β inhibition–mediated skin adverse events

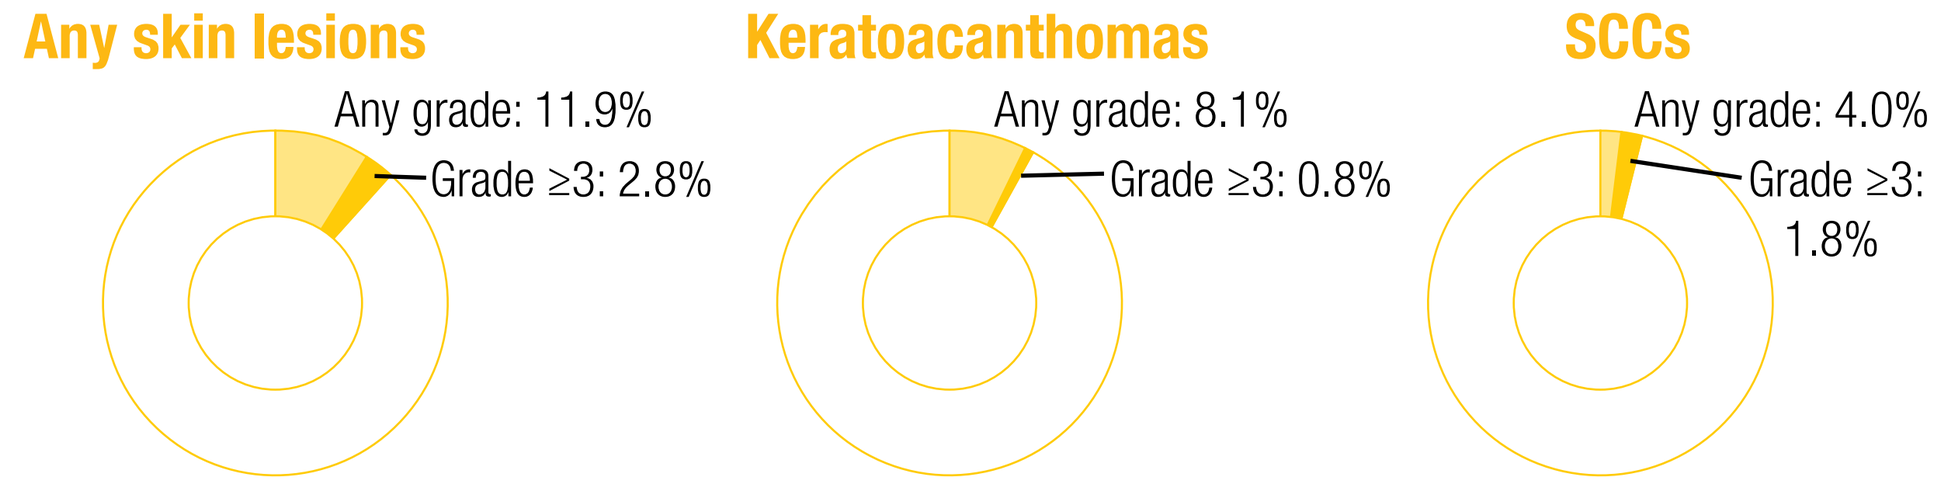
Any skin lesions
Any grade: 11.9%
Grade ≥3: 2.8%
Keratoacanthomas
Any grade: 8.1%
Grade ≥3: 0.8%
SCCs
Any grade: 4.0%
Grade ≥3: 1.8%

TGF-β inhibition–mediated skin AEs of any grade occurred in 11.9% of patients (n=72) and were generally mild or moderate. The most common TGF-β inhibition–mediated skin AEs were KAs and SCCs.

Insights: KAs occur most commonly in older light-skinned patients with a history of sun damage[3-5]

Management: refer to a dermatologist experienced with management of patients treated with ICIs. Treatment of KAs varies based on the number of lesions and level of symptomatology. For sporadic KAs, surgical excision is the standard-of-care, with alternative options including intralesional chemotherapy, cryotherapy, ablative lasers, radiotherapy, or photodynamic therapy. Observation may also be sufficient, as KAs can spontaneously regress. Treatment approaches should be based on local guidelines and standard of care. Close follow-up is recommended for re-evaluation and for monitoring of resolution and recurrence

View detailed TGF-β inhibition–mediated skin management guidance

Immune-related adverse events

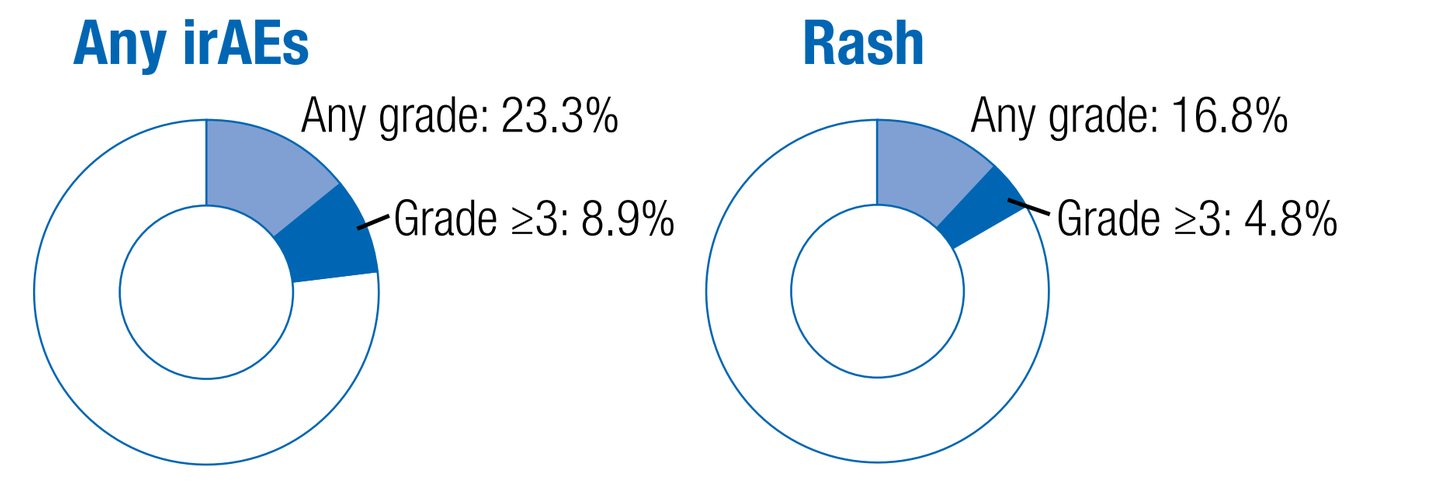
Any irAEs
Any grade: 23.3%
Grade ≥3: 8.9%
Rash
Any grade: 16.8%
Grade ≥3: 4.8%

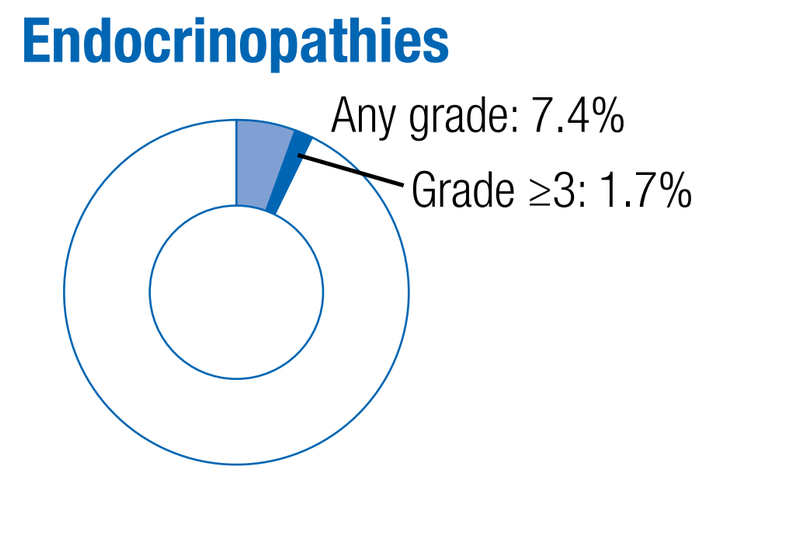
Endocrinopathies
Any grade: 7.4%
Grade ≥3: 1.7%

irAEs of any grade occurred in 23.3% of patients (n=141). The most common types of irAEs were rash and endocrinopathies. No other types of irAEs occurred in ≥5% of patients. Most irAEs were mild or moderate in severity.

Insights: the spectrum of irAEs is similar for bintrafusp alfa compared with other PD-(L)1 inhibitors

General guidance: NCCN and ASCO organ- and system-specific guidance for the management of irAEs in patients treated with ICI therapy should be followed.[6,7] Topical corticosteroids may be used for symptomatic management of rash

Management grade 1 irAEs: bintrafusp alfa may be continued with close monitoring (additional measures may be appropriate for select hematologic, neurologic and cardiac irAEs)

Management grade 2 irAEs: bintrafusp alfa should be suspended and resumed when symptoms resolve to grade ≤1. Corticosteroids (equivalent of 0.5-1 mg/kg/day of prednisone) may be administered

Management grade 3 and 4 irAEs: bintrafusp alfa should generally be suspended (grade 3) or permanently discontinued (grade 4), and high dose corticosteroids should be administered. Immunosuppressive therapy (eg, infliximab) may be required in refractory cases. For grade 4 endocrinopathies controlled with hormone replacement, treatment may continue

View detailed irAE management guidance

Anemia

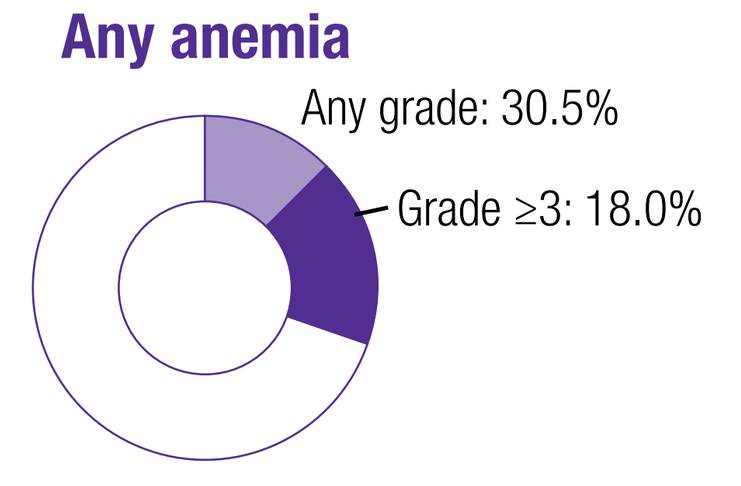
Any anemia
Any grade: 30.5%
Grade ≥3: 18.0%

Anemia of any grade occurred in 30.5% of patients (n=185). Anemia grade ≥3 occurred in 18.0% of patients (n=109).

Monitoring: hematology assessment should be performed at baseline, prior to each dose, at the end-of-treatment and 28 (±5) days post treatment

Management: anemia may be managed with standard clinical approaches, including hematologic evaluation, interventional radiology or endoscopy as appropriate. All relevant hematologic testing should be done prior to blood transfusion, if clinically feasible

View anemia management guidance

Bleeding events

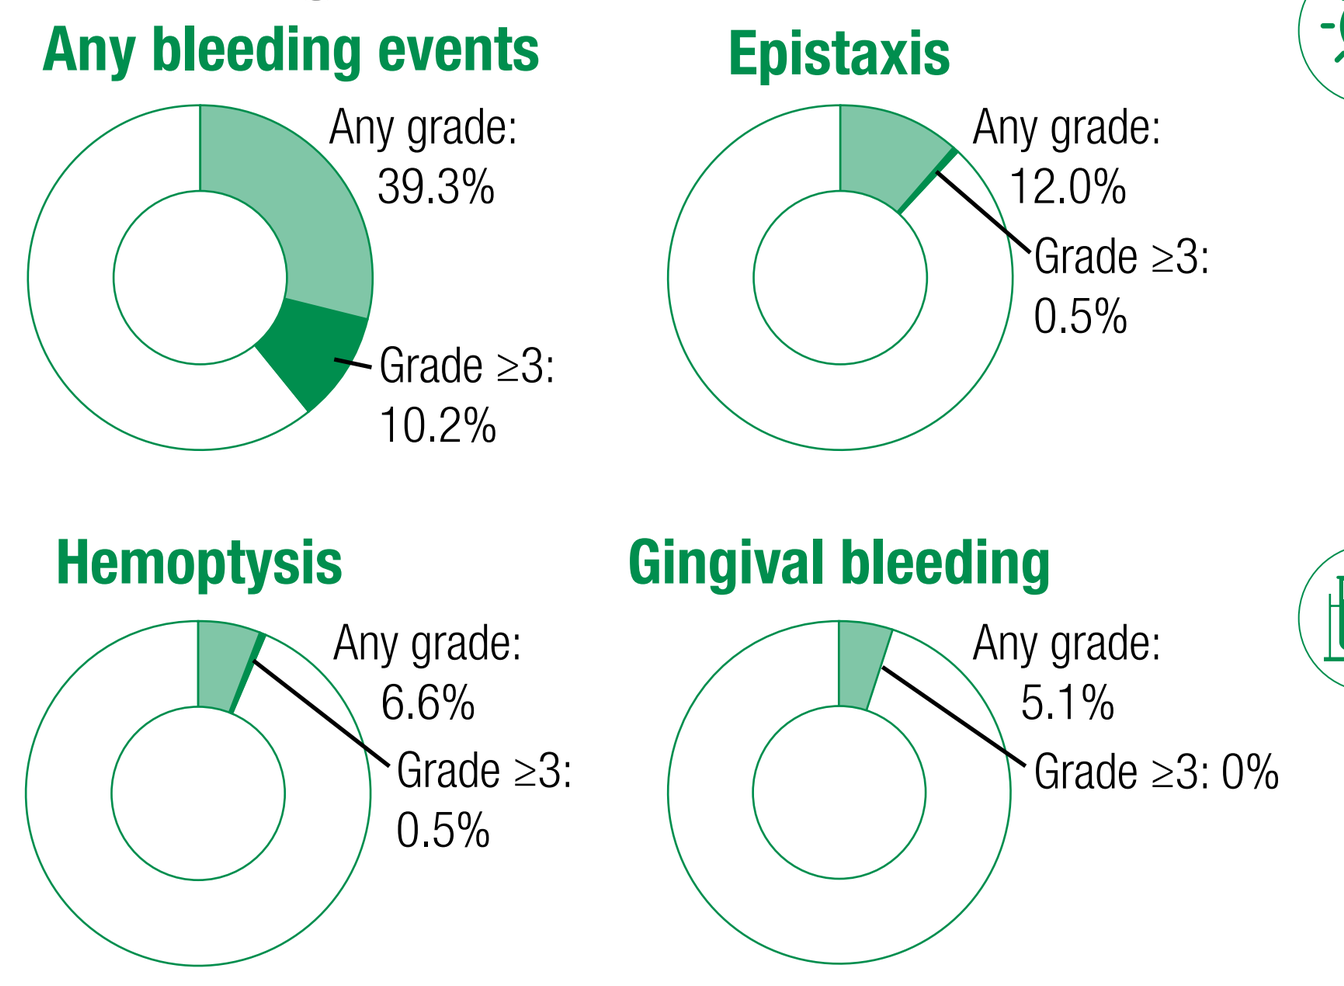
Any bleeding events
Any grade: 39.3%
Grade ≥3: 10.2%
Epistaxis
Any grade: 12.0%
Grade ≥3: 0.5%
Hemoptysis
Any grade: 6.6%
Grade ≥3: 0.5%
Gingival bleeding
Any grade: 5.1%
Grade ≥3: 0%

Insights: bleeding events may be associated with the role of TGF-β on the integrity and function of the vasculature.[8-12] Eligibility criteria for bintrafusp alfa clinical trials included an exclusion for bleeding diathesis or recent major bleeding

View management guidance for bleeding events

Management: mild to moderate mucosal bleeding events (eg, epistaxis, gingival bleeding, hematuria, hemoptysis) are clinically manageable and usually resolve without the need for bintrafusp alfa discontinuation

Bleeding events of any grade occurred in 39.3% of patients (n=238). The most common types of bleeding events (in ≥5% of patients) were epistaxis, hemoptysis,and gingival bleeding. Most bleeding events were mild or moderate in severity; the most common grade ≥3 bleeding events were GI hemorrhage (1.3%) and tumor hemorrhage (1.7%).

Infusion-related reactions

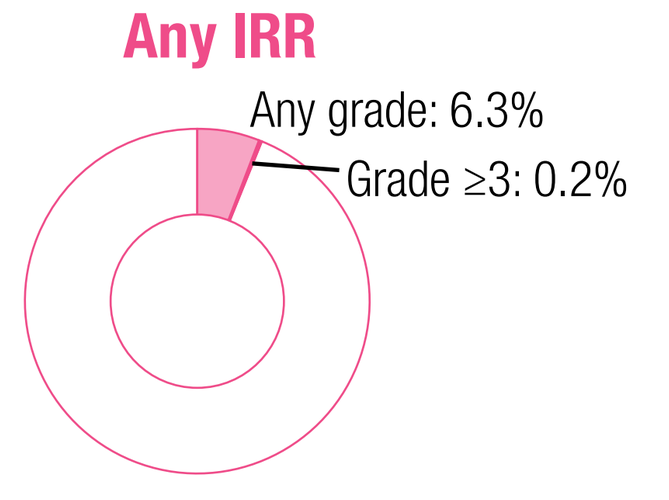
Any IRR
Any grade: 6.3%
Grade ≥3: 0.2%

IRRs occurred in 6.3% of patients (n=38). Most IRRs were mild or moderate in severity. An IRR grade ≥3 occurred in 1 patient (0.2%).

Monitoring: monitor patients on the day of and day after infusion for symptoms of hypersensitivity, including pyrexia, flushing, chills, wheezing, dyspnea, urticaria, hypotension and abdominal/back pain

Management for acute hypersensitivity/ anaphylactic reaction: care in accordance with best available medical practice should be provided, including (as applicable) epinephrine injection, intravenous dexamethasone, and cardiovascular and oxygen saturation monitoring[13]

Management for moderate IRRs: strategies may include interruption of the infusion, slowing the infusion rate, and providing symptomatic treatment

View IRR management guidance

CONCLUSION

- Most bintrafusp-alfa associated AEs were mild to moderate in severity. Timely and effective management strategies to mitigate the impact of common AEs could improve patients' QOL, time on treatment and may ultimately lead to better treatment outcomes

Abbreviations

AE, adverse event; **ASCO**, American Society of Clinical Oncology; **BTC**, biliary tract cancer; **CAF**, cancer-associated fibroblast; **EMT**, epithelial-mesenchymal transition; **GI**, gastrointestinal; **ICI**, immune checkpoint inhibitor; **irAE**, immune-related adverse event; **IRR**, infusion-related reaction; **IV**, intravenous; **KA**, keratoacanthoma; **NCCN**, National Comprehensive Cancer Network; **NK**, natural killer; **NSAID**, nonsteroidal anti-inflammatory drug; **NSCLC**, non-small cell lung cancer; **QOL**, quality of life; **SCC**, squamous cell carcinoma; **TAM**, tumor-associated macrophage; **TME**, tumor microenvironment; **TNBC**, triple-negative breast cancer; **TRAE**, treatment-related adverse event.

References

1. Strauss J, et al. Clin Cancer Res. 2018;24:1287-95.
2. Lan Y, et al. Sci Transl Med. 2018;10:eaan5488.
3. Schwartz RA, et al. J Am Acad Dermatol. 1994;30:1-19.
4. Kwiek B, Schwartz RA. J Am Acad Dermatol. 2016 Jun;74:1220-33.
5. Claeson M, et al. JAMA Dermatol. 2020 Dec 1;156:1324-1332.
6. Brahmer JR, et al. J Clin Oncol. 2018;36:1714-68.
7. NCCN Clinical Practice Guidelines in Oncology for Management of Immunotherapy-Related Toxicities V.3.2021. Accessed July 8, 2021.
8. Walshe TE, et al. PLoS One. 2009;4:e5149.
9. Cunha SI, et al. Circ Res. 2017;121:981-99.
10. Sounni NE, et al. Dis Model Mech. 2010;3:317-32.
11. Zonneville J, et al. BMC Cancer. 2018;18:670.
12. Mitra MS, et al. Toxicol Sci. 2020;175:24-34.
13. Resuscitation Council UK emergency treatment of anaphylactic reactions: guidelines for healthcare providers. Accessed July 27, 2021. https://www.resus.org.uk/pages/reaction.pdf

Acknowledgements

The authors thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers and at Merck. This study is funded by Merck (CrossRef Funder ID: 10.13039/100009945) and is part of an alliance between Merck and GlaxoSmithKline.

Medical writing support was provided by Nix Zelin, MD, of ClinicalThinking, Inc, Hamilton, NJ, USA, which was funded by Merck and GlaxoSmithKline, in accordance with Good Publication Practice (GPP3) guidelines (http://www.ismpp.org/gpp3).

Correspondence: **James L. Gulley, gulleyj@mail.nih.gov**

Disclosures

J. L. Gulley reports research funding (institution) from Merck. **M. Lacouture** reports an advisory role with Merck. **A. Spira** reports research funding from Merck and GlaxoSmithKline, and advisory roles for Merck, Bristol Myers Squibb, Mirati, Janssen, Roche, Amgen, Novartis and Sanofi. **H. Verdaguer Mata** reports an advisory role with Merck and a Speaker's Bureau role with AstraZeneca. **C. Yoo** reports research funding from Servier, Bayer, AstraZeneca, Ono Pharmaceuticals, Ipsen, Celgene and HK inno.N; an advisory role with Servier, Ipsen, AstraZeneca and Bayer; stocks/shares with OncoCross; and a Speaker's Bureau role with Merck, Servier, Bayer, AstraZeneca, Ipsen, Celgene, Merck Sharp and Dohme, HK inno.N, Debiopharm, Kyowa Kirin, Boryung Pharmaceuticals, and Janssen. **B. C. Cho** reports advisory roles for KANAPH Therapeutic Inc, Brigebio therapeutics, Cyrus therapeutics, Guardant Health; consulting roles for Novartis, AstraZeneca, Boehringer Ingelheim, Roche, Bristol Myers Squibb, Ono, Yuhan, Pfizer, Eli Lilly, Janssen, Takeda, Merck Sharp & Dohme, Medpacto, and Blueprint Medicines; research funding from Novartis, Bayer, AstraZeneca, MOGAM Institute, Dong-A ST, Champions Oncology, Janssen, Yuhan, Ono, Dizal Pharma, Merck Sharp & Dohme, AbbVie, Medpacto, GIInnovation, Eli Lilly, Blueprint Medicines, and Interpark Bio Convergence Corporation; shares/stocks for TheraCanVac, Gencurix, Bridgebio therapeutics, KANAPH Therapeutic Inc, Cyrus Therapeutics, Interpark Bio Convergence Corporation; royalties from Champions Oncology; serving on the board of directors for Interpark Bio Convergence Corp; and being the founder of Daan Biotherapeutics. **C. Helwig** reports employment with and stocks/shares from Merck. **T. Halady**, **C. Valencia**, and **M. Bajars** report employment with Merck. **J. Strauss** reports a patent (NIH) for dual PD-L1 and TGF-β inhibition in HPV-positive malignancies. All other authors have no relationships to disclose.

Abstract No. 2908. Presented at the 2021 ESMO Annual Meeting, September 17-21, 2021; E-Poster No. 1689P.