Role of prior nephrectomy for synchronous metastatic renal cell carcinoma (mRCC) on efficacy in patients treated with avelumab + axitinib or sunitinib: results from JAVELIN Renal 101

M.-O. Grimm,[1] M. Oya,[2] T. K. Choueiri,[3] M. Schmidinger,[4] D. I. Quinn,[5] G. Gravis-Mescam,[6] E. Verzoni,[7] A. J. M. Van den Eertwegh,[8] A. di Pietro,[9] M. Mariani,[9] J. Wang,[10] D. Thomaidou,[11] L. Albiges[12]

[1]Jena University Hospital, Jena, Germany; [2]Keio University School of Medicine, Tokyo, Japan; [3]Dana-Farber Cancer Institute, Boston, MA, USA; [4]Comprehensive Cancer Center, Medical University of Vienna, Vienna, Austria; [5]Keck School of Medicine USC, Los Angeles, CA, USA; [6]Institut Paoli-Calmettes, Aix-Marseille University, INSERM, CNRS, CRCM, Marseille, France; [7]Fondazione IRCCS Istituto Nazionale dei Tumori di Milano, Milan, Italy; [8]VU University Medical Center, Amsterdam, the Netherlands; [9]Pfizer SRL, Milan, Italy; [10]Pfizer, Cambridge, MA, USA; [11]Pfizer, Athens, Greece; [12]Gustave Roussy Cancer Campus, University of Paris Saclay, Villejuif, France

SCOPE

- **This analysis assessed the effect of prior nephrectomy in patients with mRCC presenting with synchronous metastases at the time of diagnosis and treated with avelumab + axitinib or sunitinib in the phase 3 JAVELIN Renal 101 trial (NCT02684006)**

CONCLUSIONS

- **In patients who presented with M1 disease at diagnosis, post hoc analyses showed that efficacy outcomes were superior in those who had undergone prior nephrectomy vs those without prior nephrectomy in the avelumab + axitinib arm but not in the sunitinib arm**
- **In the avelumab + axitinib arm, observed progression-free survival (PFS) and overall survival (OS) were numerically longer in patients with prior nephrectomy vs those without prior nephrectomy; no differences were observed between these groups in the sunitinib arm**
- **Confirmed objective response rate (ORR) was numerically higher in patients with prior nephrectomy vs those without prior nephrectomy in the avelumab + axitinib arm but not in the sunitinib arm**

GET POSTER PDF

Copies of this poster obtained through this hyperlink or quick response (QR) code are for personal use only and may not be reproduced without permission from ESMO and the author of this poster

Correspondence: **Marc-Oliver Grimm**, marc-oliver.grimm@med.uni-jena.de

GET PLAIN LANGUAGE SUMMARY

Please scan this quick response (QR) code with your smartphone app or click here to view a plain language summary of the accepted scientific abstract

BACKGROUND

- Cytoreductive nephrectomy has been a standard of care in mRCC for 20 years,[1,2] although its role in patients receiving currently available drug therapies remains controversial
 - In a randomized phase 3 trial (CARMENA), OS results showed that treatment with sunitinib alone was not inferior to cytoreductive nephrectomy followed by sunitinib[3]
 - In another randomized phase 3 trial (SURTIME), no significant difference in PFS was observed with sunitinib before deferred nephrectomy compared with cytoreductive nephrectomy followed by sunitinib[4]
- The role of prior nephrectomy in patients treated with immune checkpoint inhibitors remains unknown
- In the phase 3 JAVELIN Renal 101 trial, first-line treatment with avelumab (PD-L1 inhibitor) + axitinib (vascular endothelial growth factor receptor inhibitor) resulted in significantly longer PFS compared with sunitinib in patients with advanced RCC[5,6]
 - We assessed the effect of prior nephrectomy in patients with mRCC presenting with synchronous metastases at the time of diagnosis

METHODS

- JAVELIN Renal 101 was a multicenter, open-label, randomized phase 3 trial comparing avelumab + axitinib with sunitinib in patients with advanced RCC
- In post hoc analyses, efficacy outcomes from the third interim analysis were assessed in subgroups of patients in the avelumab + axitinib and sunitinib arms who presented with M1 disease at diagnosis and had or had not undergone prior nephrectomy
- Multivariate Cox regression analyses were used to calculate hazard ratios (HRs) for PFS (investigator assessment per RECIST 1.1) and OS
- Logistic regression method was used to calculate odds ratios for ORR (investigator assessment per RECIST 1.1)

RESULTS

- At the data cutoff (28 Apr 2020), the minimum duration of follow-up was 28 months
- 412 of 886 patients presented with M1 disease at diagnosis
 - Of these patients, 126 in the avelumab + axitinib arm and 147 in the sunitinib arm had undergone prior nephrectomy; 72 and 67, respectively, had no prior nephrectomy
- In both the avelumab + axitinib and sunitinib treatment arms, the no prior nephrectomy group had a higher proportion of patients who were older, had an impaired ECOG performance status, and had a poor risk score; however, fewer patients had PD-L1+ tumors compared with those in the prior nephrectomy group (**Table 1**)

Table 1. Baseline characteristics

	Avelumab + axitinib, n (%)		Sunitinib, n (%)	
	Prior nephrectomy (n=126)	No prior nephrectomy (n=72)	Prior nephrectomy (n=147)	No prior nephrectomy (n=67)
Age, years				
<65	86 (68.3)	46 (63.9)	100 (68.0)	34 (50.7)
≥65	40 (31.7)	26 (36.1)	47 (32.0)	33 (49.3)
Sex				
Male	91 (72.2)	51 (70.8)	115 (78.2)	46 (68.7)
Female	35 (27.8)	21 (29.2)	32 (21.8)	21 (31.3)
ECOG performance status				
0	80 (63.5)	32 (44.4)	85 (57.8)	31 (46.3)
1	46 (36.5)	40 (55.6)	62 (42.2)	35 (52.2)
2	0	0	0	1 (1.5)
IMDC prognostic criteria				
Favorable	8 (6.3)	2 (2.8)	7 (4.8)	0
Intermediate	96 (76.2)	41 (56.9)	116 (78.9)	40 (59.7)
Poor	22 (17.5)	29 (40.3)	23 (15.6)	27 (40.3)
Unknown	0	0	1 (0.7)	0
MSKCC prognostic risk group				
Favorable	8 (6.3)	2 (2.8)	10 (6.8)	0
Intermediate	103 (81.7)	43 (59.7)	121 (82.3)	47 (70.1)
Poor	14 (11.1)	27 (37.5)	15 (10.2)	20 (29.9)
Unknown	1 (0.8)	0	1 (0.7)	0
Pooled geographic region				
Europe	47 (37.3)	25 (34.7)	62 (42.2)	36 (53.7)
North America	45 (35.7)	22 (30.6)	45 (30.6)	15 (22.4)
Asia	21 (16.7)	19 (26.4)	25 (17.0)	11 (16.4)
Rest of the world	13 (10.3)	6 (8.3)	15 (10.2)	5 (7.5)
PD-L1 status				
Positive	96 (76.2)	29 (40.3)	114 (77.6)	26 (38.8)
Negative	26 (20.6)	30 (41.7)	27 (18.4)	29 (43.3)
Unknown	4 (3.2)	13 (18.1)	6 (4.1)	12 (17.9)

IMDC, International Metastatic RCC Database Consortium; **MSKCC**, Memorial Sloan Kettering Cancer Center.

- After adjusting for imbalances in different variables:
 - In the avelumab + axitinib arm, the risk of progression or death was numerically lower in the prior nephrectomy group than in the no prior nephrectomy group (HR, 0.785; 95% CI, 0.531-1.161); in contrast, in the sunitinib arm, the risk of progression or death was numerically higher in the prior nephrectomy group than in the no prior nephrectomy group (HR, 1.146; 95% CI, 0.773-1.699) (**Table 2**)
 - In the avelumab + axitinib arm, the risk of death was numerically lower in the prior nephrectomy group than in the no prior nephrectomy group (HR, 0.593; 95% CI, 0.379-0.930); in the sunitinib arm, no difference was observed between the groups (HR, 0.859; 95% CI, 0.551-1.341) (**Table 3**)
 - In the avelumab + axitinib arm, the odds of response was numerically higher in the prior nephrectomy group than in the no prior nephrectomy group (HR, 2.669; 95% CI, 1.315-5.414), whereas in the sunitinib arm, no difference was observed between the groups (HR, 2.018; 95% CI, 0.824-4.941) (**Table 4**)

Table 2. Multivariate Cox regression analysis for PFS based on investigator assessment per RECIST 1.1

Variables	Levels	Parameter estimate	Standard error	2-sided p value	HR (95% CI)
Prior nephrectomy	1: Yes	0.14	0.20	0.4976	
	0: No				
Treatment	1: Avelumab + axitinib	–0.19	0.23	0.4048	
	0: Sunitinib				
Age, years	1: ≥65	–0.24	0.13	0.0601	
	0: <65				
Pooled geographic region	1: North America	0.52	0.18	0.0051	
	2: Europe	0.17	0.18	0.3498	
	3: Rest of the world	0.13	0.25	0.6077	
	0: Asia				
ECOG performance status	1: 1	0.20	0.13	0.1258	
	2: 2	4.54	1.24	0.0003	
	0: 0				
IMDC prognostic criteria	1: Intermediate	0.50	0.32	0.1154	
	2: Poor	0.89	0.34	0.0087	
	0: Favorable				
PD-L1 status	1: Positive	0.13	0.14	0.3683	
	0: Negative				
Prior nephrectomy × treatment		–0.38	0.27	0.1641	
Prior nephrectomy: yes vs no for avelumab + axitinib					0.785 (0.531-1.161)
Prior nephrectomy: yes vs no for sunitinib					1.146 (0.773-1.699)

HR, hazard ratio; **IMDC**, International Metastatic RCC Database Consortium; **PFS**, progression-free survival.

Table 3. Multivariate Cox regression analysis for OS

Variables	Levels	Parameter estimate	Standard error	2-sided p value	HR (95% CI)
Prior nephrectomy	1: Yes	–0.15	0.23	0.5043	
	0: No				
Treatment	1: Avelumab + axitinib	–0.04	0.25	0.8646	
	0: Sunitinib				
Age, years	1: ≥65	–0.13	0.15	0.4026	
	0: <65				
Pooled geographic region	1: North America	0.45	0.23	0.0488	
	2: Europe	0.34	0.23	0.1392	
	3: Rest of the world	0.11	0.33	0.7448	
	0: Asia				
ECOG performance status	1: 1	0.53	0.15	0.0005	
	2: 2	5.36	1.43	0.0002	
	0: 0				
IMDC prognostic criteria	1: Intermediate	0.83	0.51	0.1039	
	2: Poor	1.34	0.52	0.0106	
	0: Favorable				
PD-L1 status	1: Positive	0.19	0.17	0.2611	
	0: Negative				
Prior nephrectomy × treatment		–0.37	0.31	0.2256	
Prior nephrectomy: yes vs no for avelumab + axitinib					0.593 (0.379-0.930)
Prior nephrectomy: yes vs no for sunitinib					0.859 (0.551-1.341)

HR, hazard ratio; **IMDC**, International Metastatic RCC Database Consortium; **OS**, overall survival.

Table 4. Logistic regression analysis for confirmed ORR based on investigator assessment per RECIST 1.1

Variables	Levels	Parameter estimate	Standard error	2-sided p value	Odds ratio (95% CI)
Prior nephrectomy	1: Yes	0.70	0.46	0.1244	
	0: No				
Treatment	1: Avelumab + axitinib	1.35	0.50	0.0064	
	0: Sunitinib				
Age, years	1: ≥65	0.28	0.25	0.2669	
	0: <65				
Pooled geographic region	1: North America	–1.05	0.35	0.0025	
	2: Europe	–0.94	0.33	0.0046	
	3: Rest of the world	–0.80	0.46	0.0836	
	0: Asia				
ECOG performance status	1: 1	–0.23	0.25	0.3619	
	2: 2	–10.51	759.71	0.9890	
	0: 0				
IMDC prognostic criteria	1: Intermediate	–0.20	0.56	0.7227	
	2: Poor	–1.15	0.62	0.0630	
	0: Favorable				
PD-L1 status	1: Positive	–0.03	0.28	0.9065	
	0: Negative				
Prior nephrectomy × treatment		0.28	0.56	0.6202	
Prior nephrectomy: yes vs no for avelumab + axitinib					2.669 (1.315-5.414)
Prior nephrectomy: yes vs no for sunitinib					2.018 (0.824-4.941)

IMDC, International Metastatic RCC Database Consortium; **ORR**, objective response rate.

REFERENCES 1. Flanigan RC, et al. J Urol. 2004;171(3):1071-6. **2.** Escudier B, et al. Ann Oncol. 2016;27(suppl 5):v58-68. **3.** Méjean A, et al. N Engl J Med. 2018;379(5):417-27. **4.** Bex A, et al. JAMA Oncol. 2019;5(2):164-70. **5.** Motzer RJ, et al. N Engl J Med. 2019;380(12):1103-15. **6.** Choueiri TK, et al. Ann Oncol. 2020;31(8):1030-9.

DISCLOSURES M.-O. Grimm has received honoraria from Astellas Pharma, AstraZeneca, Bristol Myers Squibb, EUSA Pharma, Ipsen, Merck, MSD, and Pfizer; has served in a consulting or advisory role for Astellas Pharma, AstraZeneca, Bristol Myers Squibb, Eisai, EUSA Pharma, Merck, MSD, Pfizer, Roche Pharma AG, and Takeda; has received research funding, paid to his institution, from Bristol Myers Squibb and Intuitive; and has received travel, accommodations, and expenses from Bristol Myers Squibb and Merck. **M. Oya** has provided speaker services for Bayer, Bristol Myers Squibb, MSD, Novartis, Ono, Pfizer, and Takeda. **T. K. Choueiri** holds stock and additional ownership interests in Pionyr and Tempest Therapeutics; has received honoraria from Alexion Pharmaceuticals, Alligent, Analysis Group, American Society of Clinical Oncology (ASCO), AstraZeneca, Bayer, Bristol Myers Squibb, Cerulean Pharma, Clinical Care Options, Corvus Pharmaceuticals, Eisai, EMD Serono, Exelixis, Foundation Medicine, Genentech/Roche, GlaxoSmithKline, Harborside Press, HERON, Ipsen, Kidney Cancer Association, Lancet Oncology, Lilly, Lpath, Michael J. Hennessy Associates, MSD, Navinata Health, National Comprehensive Cancer Network (NCCN), Novartis, Peloton Therapeutics, Pfizer, PlatformQ Health, Prometheus, Sanofi, New England Journal of Medicine, and UpToDate; has served in a consulting or advisory role for Alexion Pharmaceuticals, Alligent, Analysis Group, ASCO, AstraZeneca, Bayer, Bristol Myers Squibb, Cerulean Pharma, Clinical Care Options, Corvus Pharmaceuticals, Eisai, EMD Serono, European Society for Medical Oncology (ESMO), Exelixis, Foundation Medicine, GlaxoSmithKline, Harborside Press, HERON, Ipsen; Kidney Cancer Association, Lancet Oncology, Lilly, Lpath, Michael J. Hennessy Associates, MSD, Navinata Health, NCCN, Novartis, Peloton Therapeutics, Pfizer, PlatformQ Health, Prometheus, Roche/Genentech, Sanofi, New England Journal of Medicine, and UpToDate; has received research funding, paid to his institution, from Agensys, Analysis Group, AstraZeneca, Bayer, Bristol Myers Squibb, Calithera Biosciences, Celldex, Cerulean Pharma, Congressionally Directed Medical Research Programs (DOD), Corvus Pharmaceuticals, Eisai, Exelixis, Foundation Medicine, Gateway for Cancer Research, GlaxoSmithKline, Ipsen, MSD, NCI, Novartis, Peloton Therapeutics, Pfizer, Prometheus, Roche, Roche/Genentech, Seattle Genetics/Astellas Pharma, Takeda, and TRACON Pharmaceuticals; holds patents, royalties, or other intellectual property for international patent application no. PCT/US2018/058430, entitled "Biomarkers of Clinical Response and Benefit to Immune Checkpoint Inhibitor Therapy" and international patent application no. PCT/US2018/12209, entitled "PBRM1 Biomarkers Predictive of Anti-Immune Checkpoint Response"; and has received travel, accommodations, and expenses from Alexion Pharmaceuticals, Alligent, Analysis Group, AstraZeneca, Bayer, Bristol Myers Squibb, Cerulean Pharma, Clinical Care Options, Corvus Pharmaceuticals, Eisai, EMD Serono, ESMO, Exelixis, Foundation Medicine, GlaxoSmithKline, Harborside Press, HERON, Ipsen, Kidney Cancer Association, Lancet Oncology, Lilly, Lpath, Michael J. Hennessy Associates, MSD, Navinata Health, NCCN, Novartis, Peloton Therapeutics, Pfizer, PlatformQ Health, Prometheus, Roche/Genentech, Sanofi, New England Journal of Medicine, and UpToDate. **M. Schmidinger** has served in advisory role for Alkermes, Bristol Myers Squibb, Eisai, EUSA Pharma, Exelixis, Ipsen, Merck, MSD, Pfizer, and Roche; has received honoraria from Bristol Myers Squibb, Eisai, EUSA, Exelixis, Ipsen, Merck, MSD, and Pfizer. **D. I. Quinn** has received research grant from Genentech, MSD, and Pfizer; has served in an advisory role for Astellas Pharma, AstraZeneca, Bayer, Bristol Myers Squibb, EMD Serono, Exelixis, Genentech, MSD, Novartis, Pfizer, and Roche; holds stocks and has provided speaker services for NIL; and has participated in data safety monitoring for Eisai and US Biotest. **G. Gravis-Mescam** has received reimbursement for travel and accommodations expenses from Astellas Pharma, Bristol Myers Squibb, Ipsen, Janssen Oncology, Pfizer, and Sanofi. **E. Verzoni** has served in advisory role for Ipsen, Merck, MSD, and Pfizer. **A. J. M. Van den Eertwegh** has received research funding, paid to his institution, from Roche, Sanofi, and TEVA; has served in advisory role, paid to him and his institution, for Amgen, Bristol Myers Squibb, Eisai, Ipsen, Merck & Co, MSD, Novartis, Pfizer, and Pierre Fabre; and has provided speaker services for Bristol Myers Squibb. **A. di Pietro** is an employee and holds stock at Pfizer. **M. Mariani** is an employee and holds stock at Pfizer. **J. Wang** is an employee at Pfizer. **D. Thomaidou** is an employee and holds stock at Pfizer. **L. Albiges** has received research funding, paid to his institution, from Bristol Myers Squibb; has served in advisory role, for his institution, for Astellas Pharma, AstraZeneca, Bellerophon, Bristol Myers Squibb, Corvus Pharmaceuticals, Ipsen, Janssen, Merck, MSD, Novartis, Pfizer, and Springer Healthcare. **ACKNOWLEDGMENTS** The authors would like to thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers. This trial was sponsored by Pfizer as part of an alliance between Pfizer and Merck (CrossRef Funder ID: 10.13039/100009945). Medical writing support was provided by Kakoli Parai of ClinicalThinking and funded by Pfizer and Merck.

Poster No. 665P. Presented at the ESMO Congress 2021, 16-21 September, 2021; Virtual.